The FORCE™ Platform Enables TfR1-Mediated Delivery of Exon Skipping PMO to the CNS and Resolves Anxiety in a Mouse Model of DMD

Cody A. Desjardins, Si Han Li, John Hall, Reshmii Venkatesan, Susana Correia, Jin Cui, Jennifer Johnson, Esther Aix, Tim Weeden, Oxana Beskrovnaya, Stefano Zanotti

Dyne Therapeutics, Inc., Waltham, MA, USA

INTRODUCTION

Neuropsychiatric symptoms are an unmet need in DMD. Many people with DMD experience central nervous system (CNS) symptoms, including anxiety, attention-deficit disorder, and learning disabilities, driven by CNS deficits consistent with loss of dystrophin in the brain[1,2]. While therapies for DMD leveraging phosphorodiamidate morpholino oligomers (PMO) induce exon skipping in the *DMD* pre-mRNA and enable translation of an internally-truncated near full-length dystrophin, lack of distribution to the brain precludes addressing the CNS manifestations of DMD[3].

We developed the FORCE™ platform to enhance uptake of diverse therapeutic payloads in muscle. We have previously demonstrated that our platform delivers charged oligonucleotides and protein payloads to the CNS[4]. Preclinical data in *mdx* mice and initial results from the DYNE-251 clinical program in exon 51 skipping amenable DMD patients showed that the FORCE platform enhances PMO delivery and efficacy in muscle[5]; however, its ability to do so in CNS remains to be demonstrated.

Mdx mice exhibit anxiety-induced freezing behavior in response to restraint[6]. To evaluate whether the FORCE platform can correct this phenotype, we generated the hTfR1/*mdx* mouse model expressing human TFR1. The hTfR1/*mdx* mouse recapitulates the anxiety-induced freezing behavior of the parental *mdx* strain. This model allowed us to test the efficacy of a human TfR1-targeting Fab and PMO conjugate designed to skip exon 23 of murine *Dmd* pre-mRNA (FORCE-M23D). A single intravenous dose of FORCE-M23D, but not unconjugated M23D or non-TfR1 targeting negative control conjugate, led to widespread PMO distribution to CNS in hTfR1/*mdx* mice. PMO delivery to CNS induced *Dmd* pre-mRNA exon skipping and restoration of dystrophin protein expression, leading to complete and durable resolution of anxiety-induced freezing behavior. To our knowledge, this is the only platform with demonstrated potential for a systemically-administered therapy to restore dystrophin protein in the brain.

Figure 1. Dyne FORCE Platform Leverage TfR1 To Deliver Payloads to the CNS

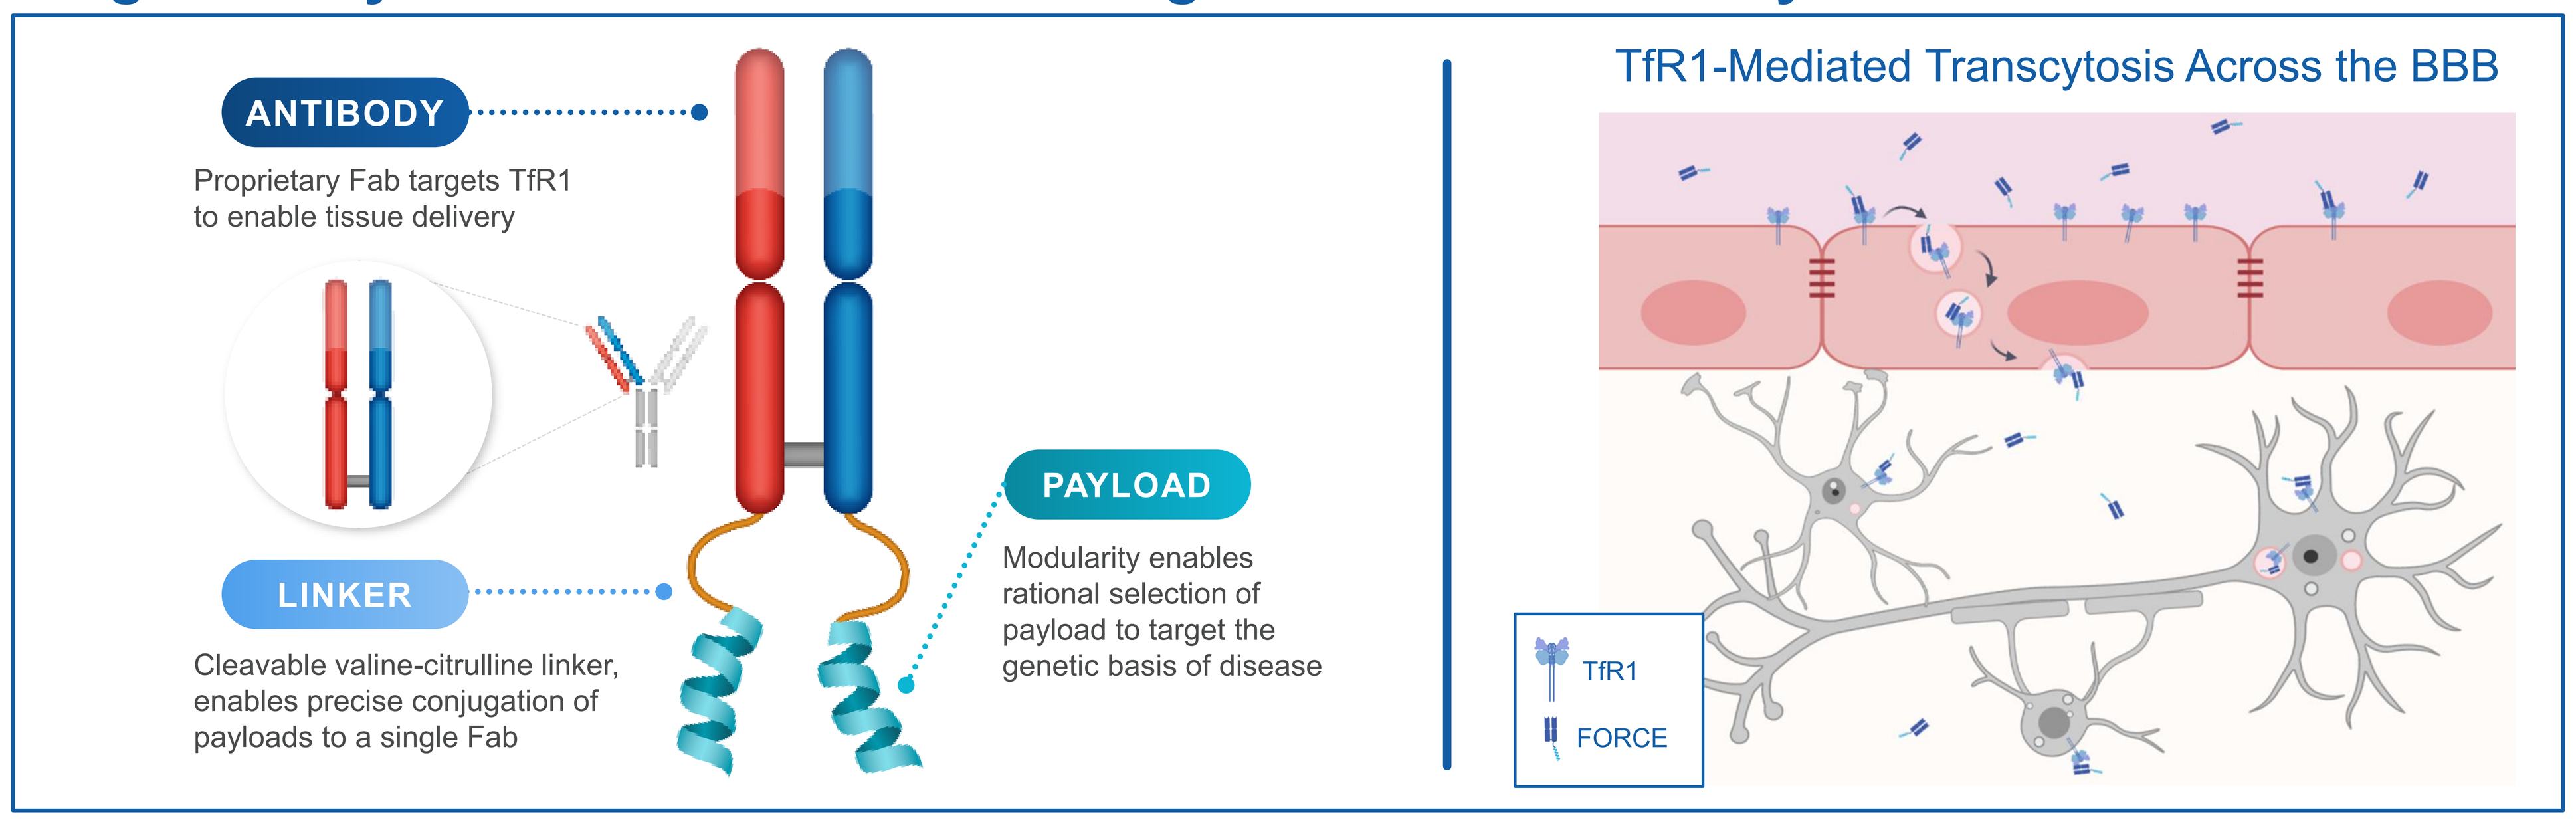
ANTIBODY
Proprietary Fab targets TfR1 to enable tissue delivery
LINKER
Cleavable valine-citrulline linker, enables precise conjugation of payloads to a single Fab
PAYLOAD
Modularity enables rational selection of payload to target the genetic basis of disease
TfR1-Mediated Transcytosis Across the BBB
TfR1
FORCE

Figure 2. hTfR1/*mdx* Mouse Model Enables Assessment of FORCE Delivery of PMO to the CNS

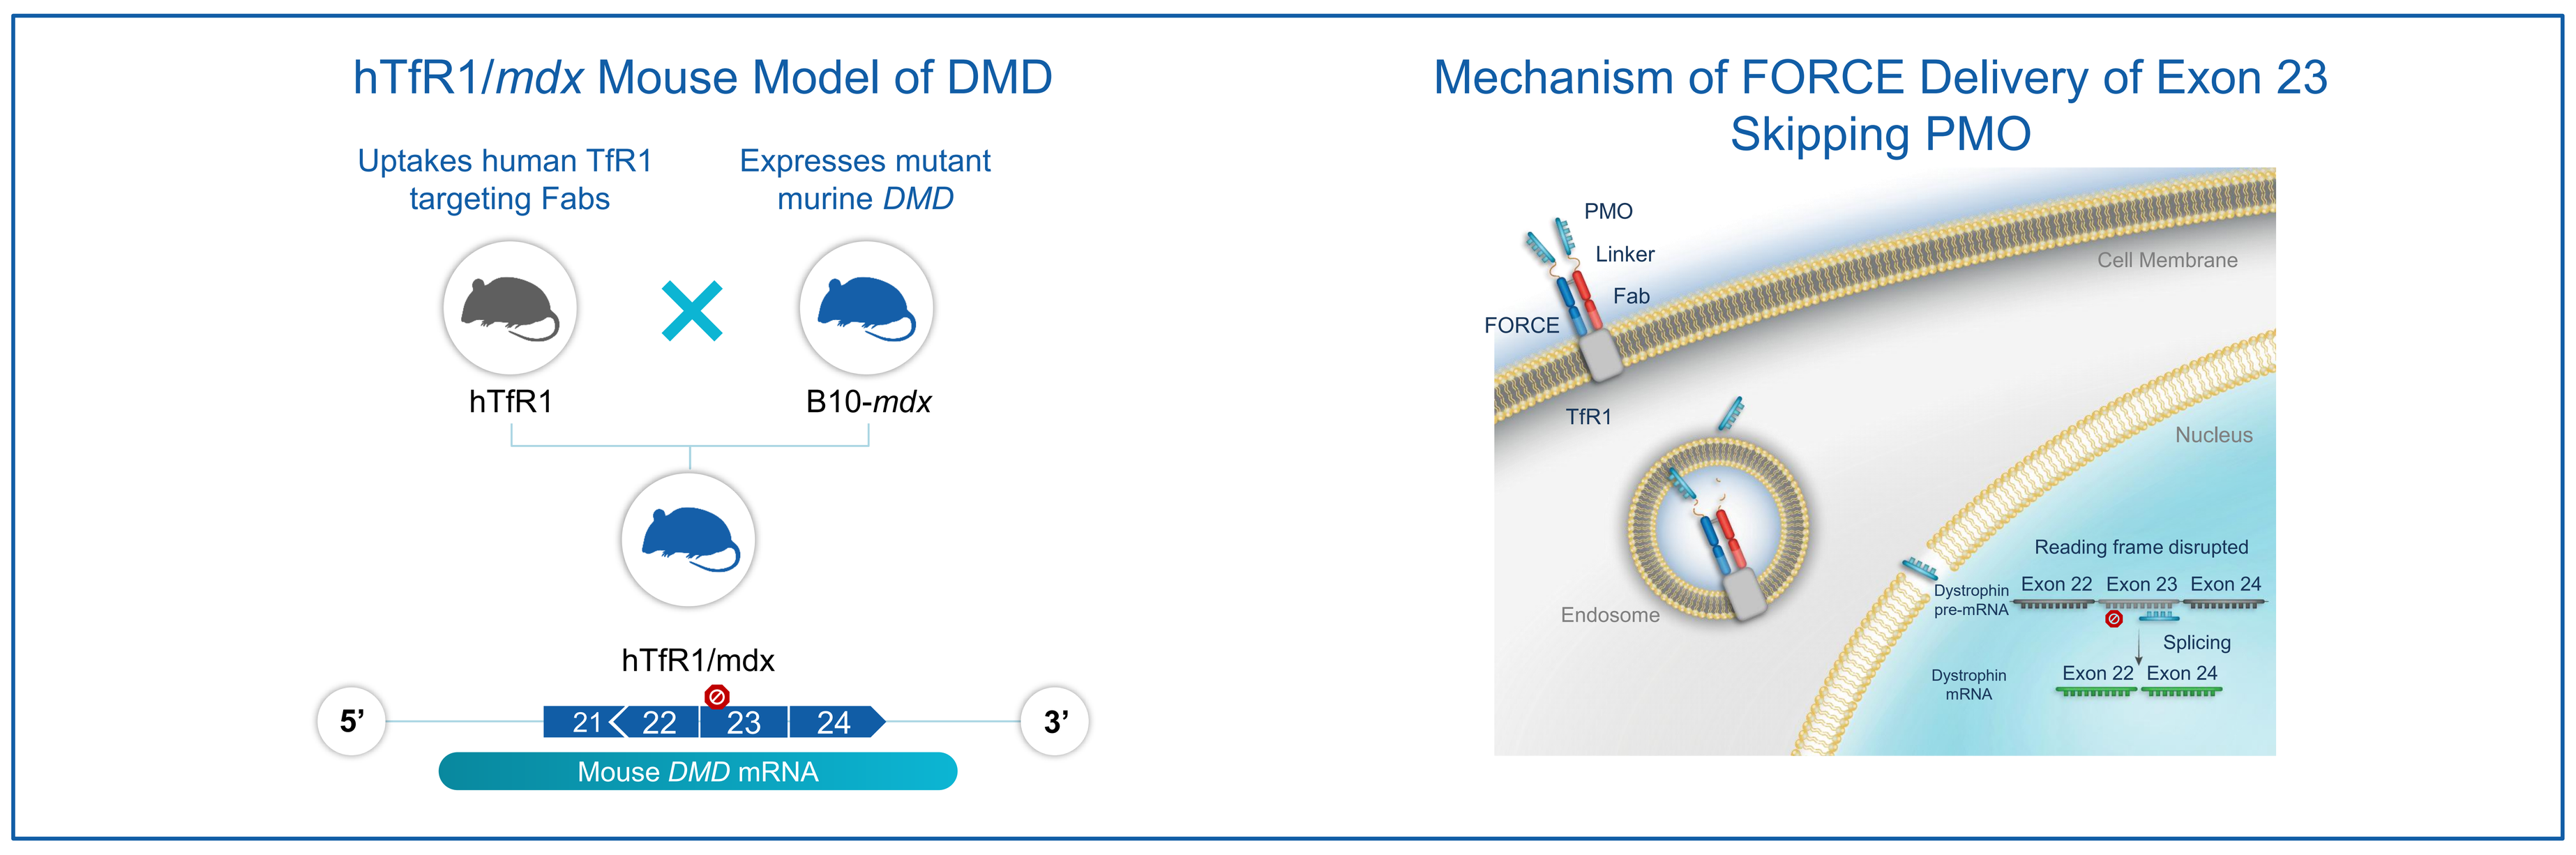
hTfR1/mdx Mouse Model of DMD
Uptakes human TfR1 targeting Fabs
Expresses mutant murine DMD
hTfR1
B10-mdx
hTfR1/mdx
5'
21
22
23
24
3'
Mouse DMD mRNA
Mechanism of FORCE Delivery of Exon 23 Skipping PMO
PMO
Linker
Fab
FORCE
TfR1
Cell Membrane
Nucleus
Endosome
Reading frame disrupted
Dystrophin pre-mRNA
Exon 22
Exon 23
Exon 24
Splicing
Dystrophin mRNA

Figure 3. Schematic Representation of Restraint-Induced Behavioral Testing

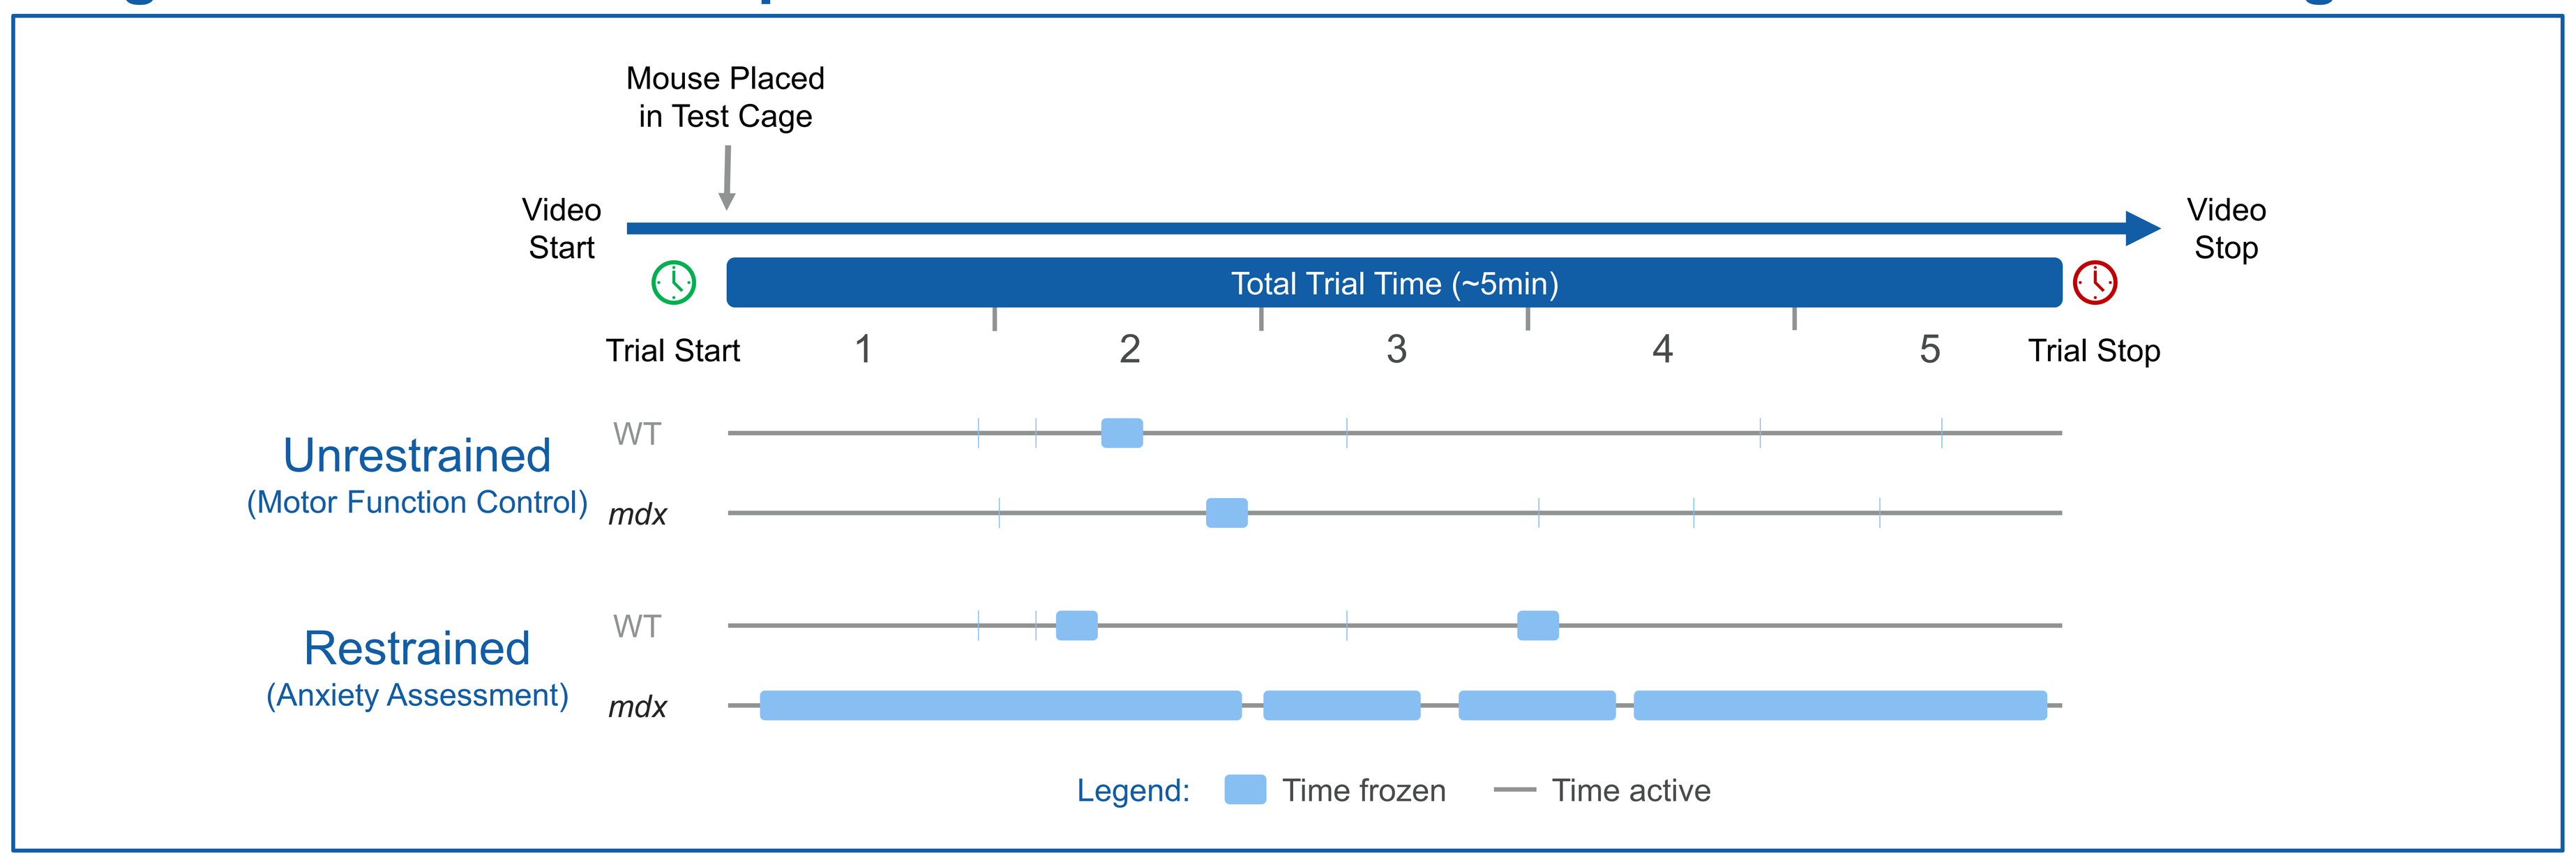
Mouse Placed in Test Cage
Video Start
Video Stop
Total Trial Time (~5min)
Trial Start
1
2
3
4
5
Trial Stop
Unrestrained
(Motor Function Control)
Restrained
(Anxiety Assessment)
WT
mdx
Legend:
Time frozen
Time active

Restraint-induced freezing was assessed in WT or *mdx* mice restrained by scruffing for ~15-20 seconds. Animals were then placed in a fresh cage and a 5 minutes video was captured. Freezing was quantified in five 1-minute segments beginning when the animal is placed in the cage and % time frozen was averaged across the total trial.

RESULTS

Figure 4. FORCE-M23D Delivers PMO Across the BBB into the Brain Parenchyma

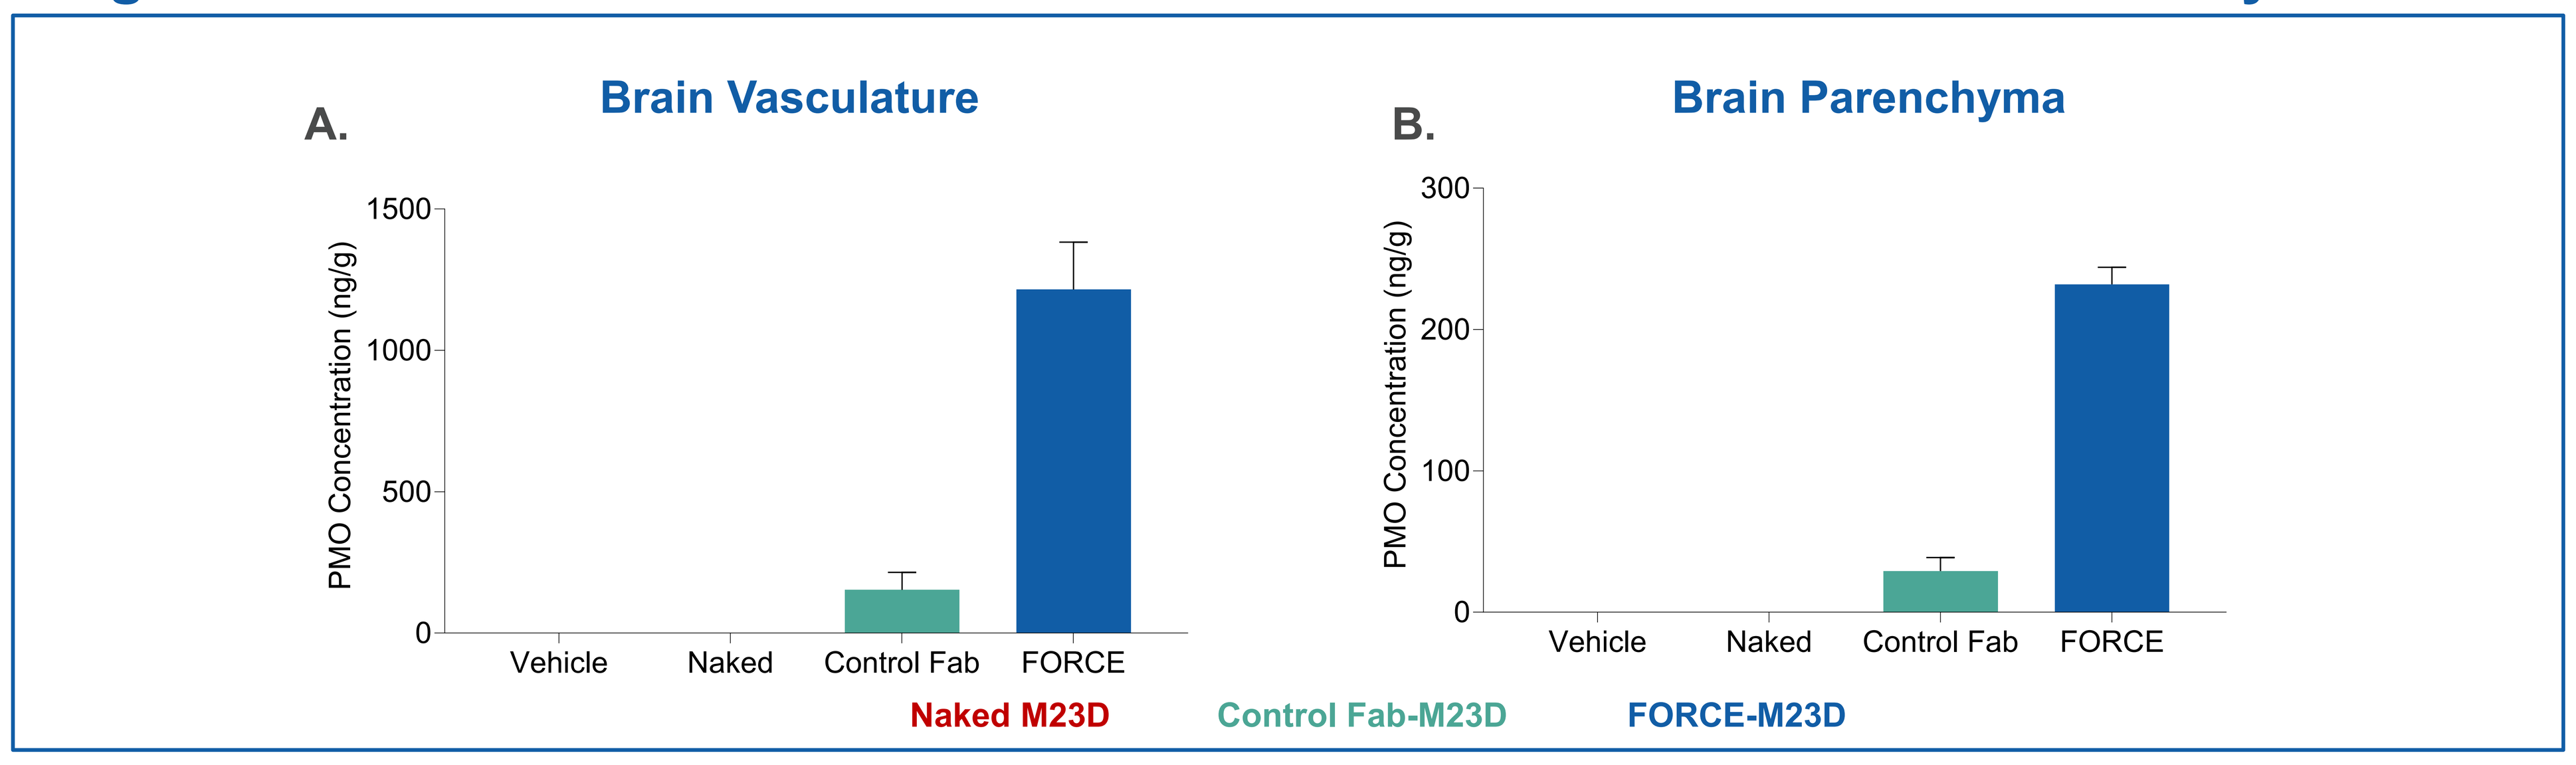
A.
Brain Vasculature
PMO Concentration (ng/g)
1500
1000
500
0
Vehicle
Naked
Control Fab
FORCE
B.
Brain Parenchyma
300
200
100
Naked M23D
Control Fab-M23D
FORCE-M23D

FORCE-M23D shows successful delivery to the brain vasculature (A) and parenchyma (B) compared to naked M23D or M23D conjugated to a non-targeting negative control Fab. . Male *mdx* mice were administered a single tail vein IV dose of vehicle or 30 mg/kg equivalent of test articles and a hemisphere of the brain was collected 7 days post-dose. Data are mean ±SD, n= 5 mice/group. Statistical significance is one-way ANOVA with a Tukey's HSD post-hoc test comparing all groups; **** p<0.0001.

Figure 5. FORCE-M23D Delivers Superior PMO Exposure and Exon Skipping 7 Days After a Single IV Administration in hTfR1/*mdx* Mice

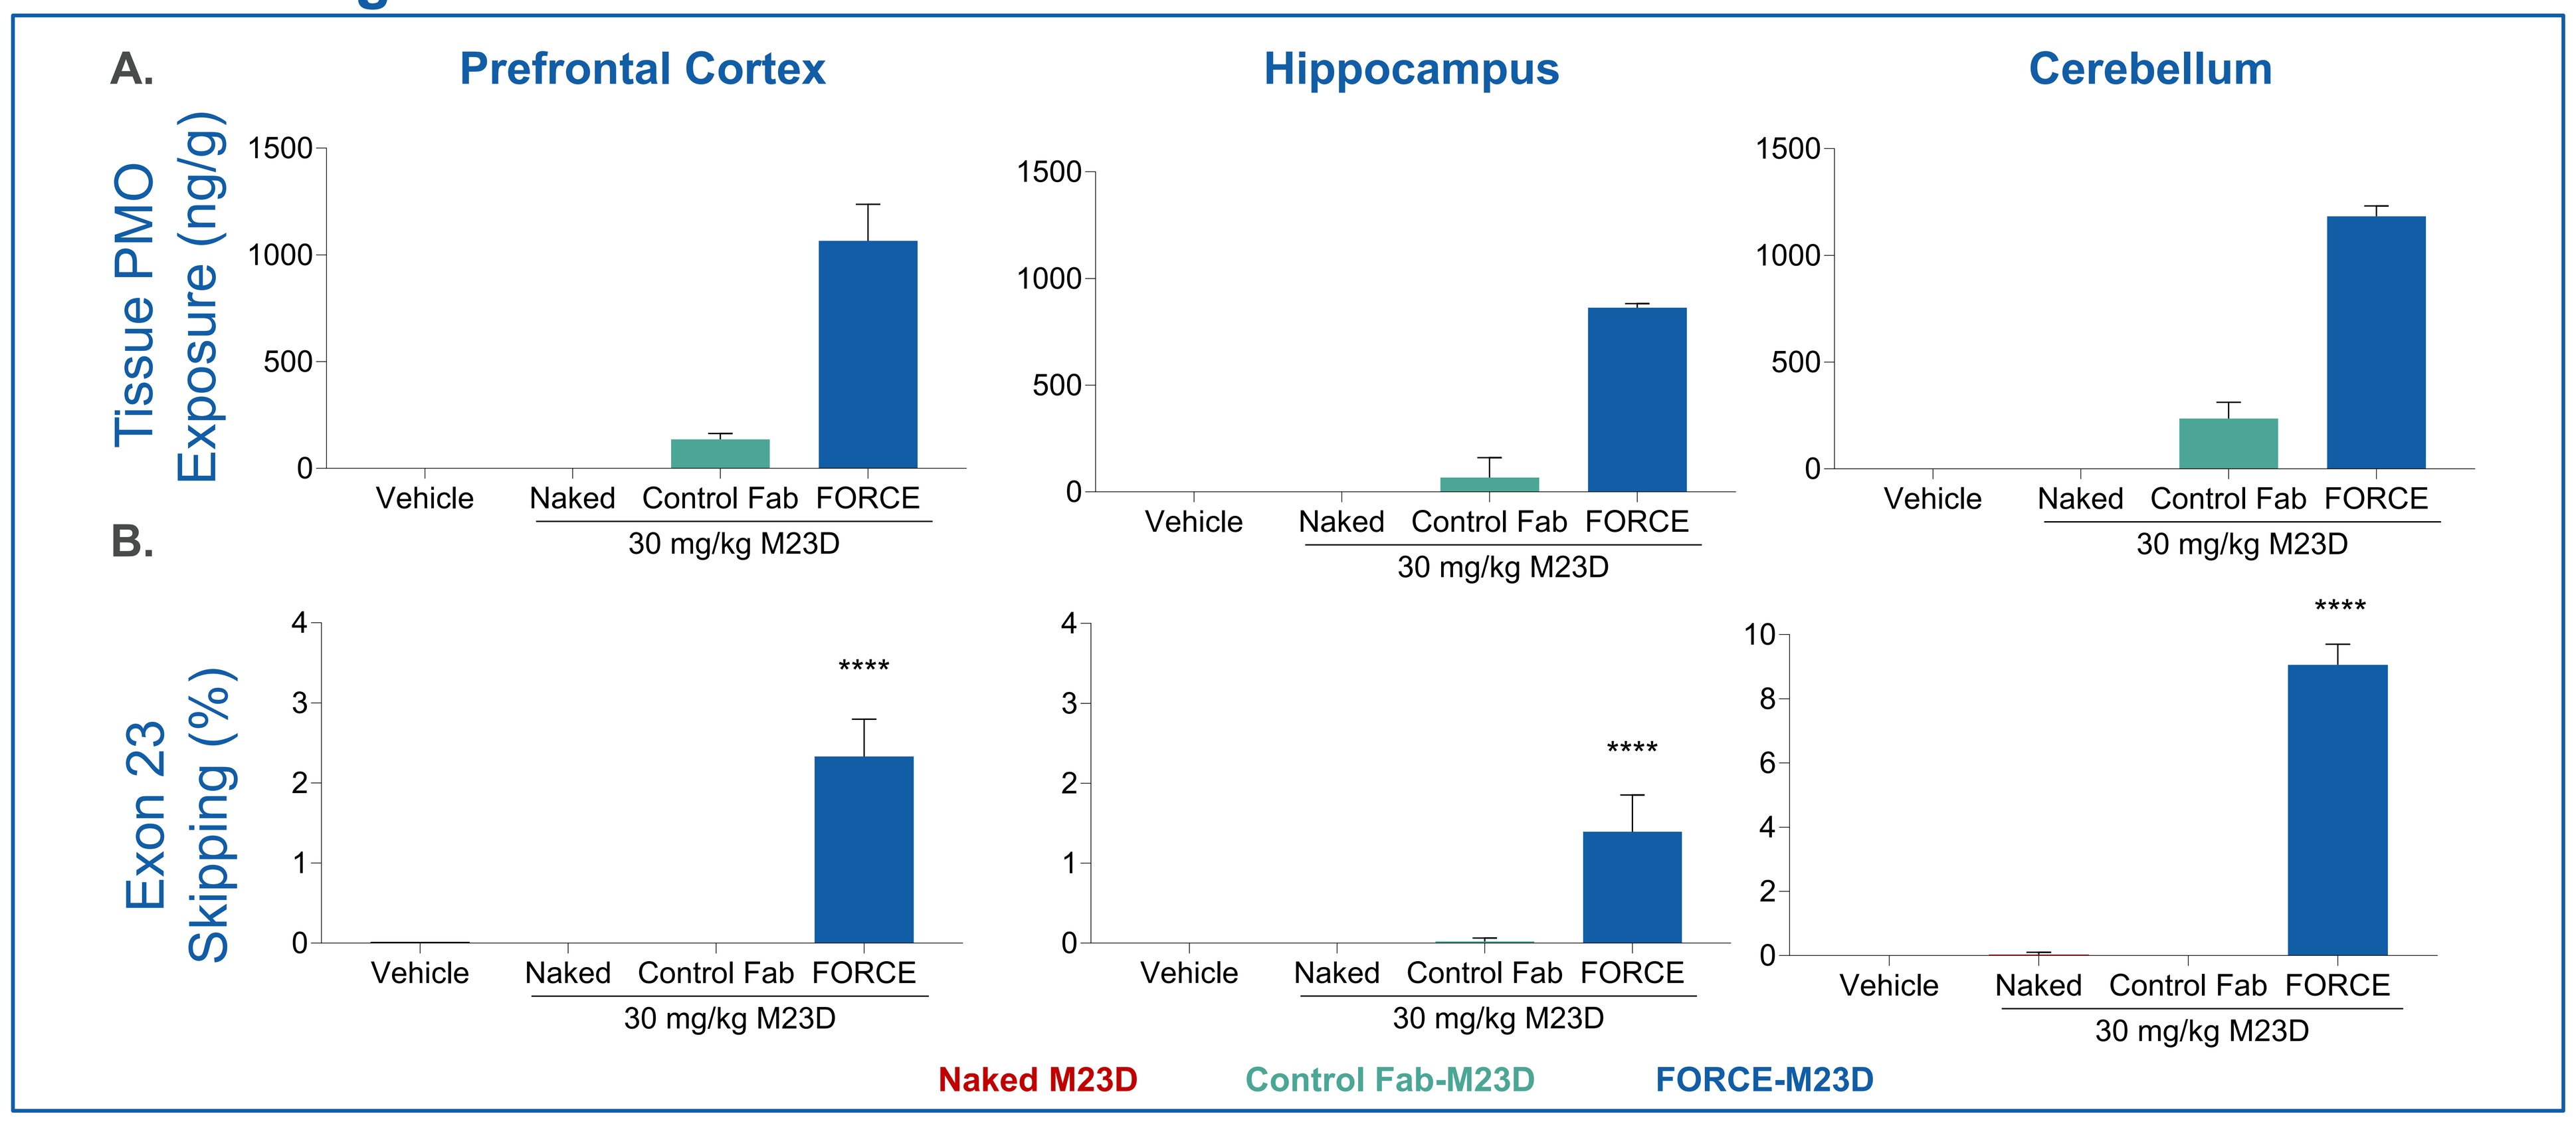
A.
Prefrontal Cortex
Hippocampus
Cerebellum
Tissue PMO Exposure (ng/g)
1500
1000
500
0
Vehicle
Naked
Control Fab
FORCE
30 mg/kg M23D
B.
Exon 23 Skipping (%)

Naked M23D
Control Fab-M23D
FORCE-M23D

FORCE-M23D shows superior delivery (A) and pharmacology (B) in the CNS of *mdx* mice compared to naked M23D or M23D conjugated to a non-targeting negative control Fab. Male *mdx* mice were administered a single tail vein IV dose of vehicle or 30 mg/kg equivalent of test articles and tissues were collected 7 days post-dose. Data are mean ±SD, n= 5 mice/group. Statistical significance is one-way ANOVA with a Tukey's HSD post-hoc test comparing all groups; **** p<0.0001.

Figure 6. hTfR1/*mdx* Mice Exhibit Durable Freezing Behavior After Restraint

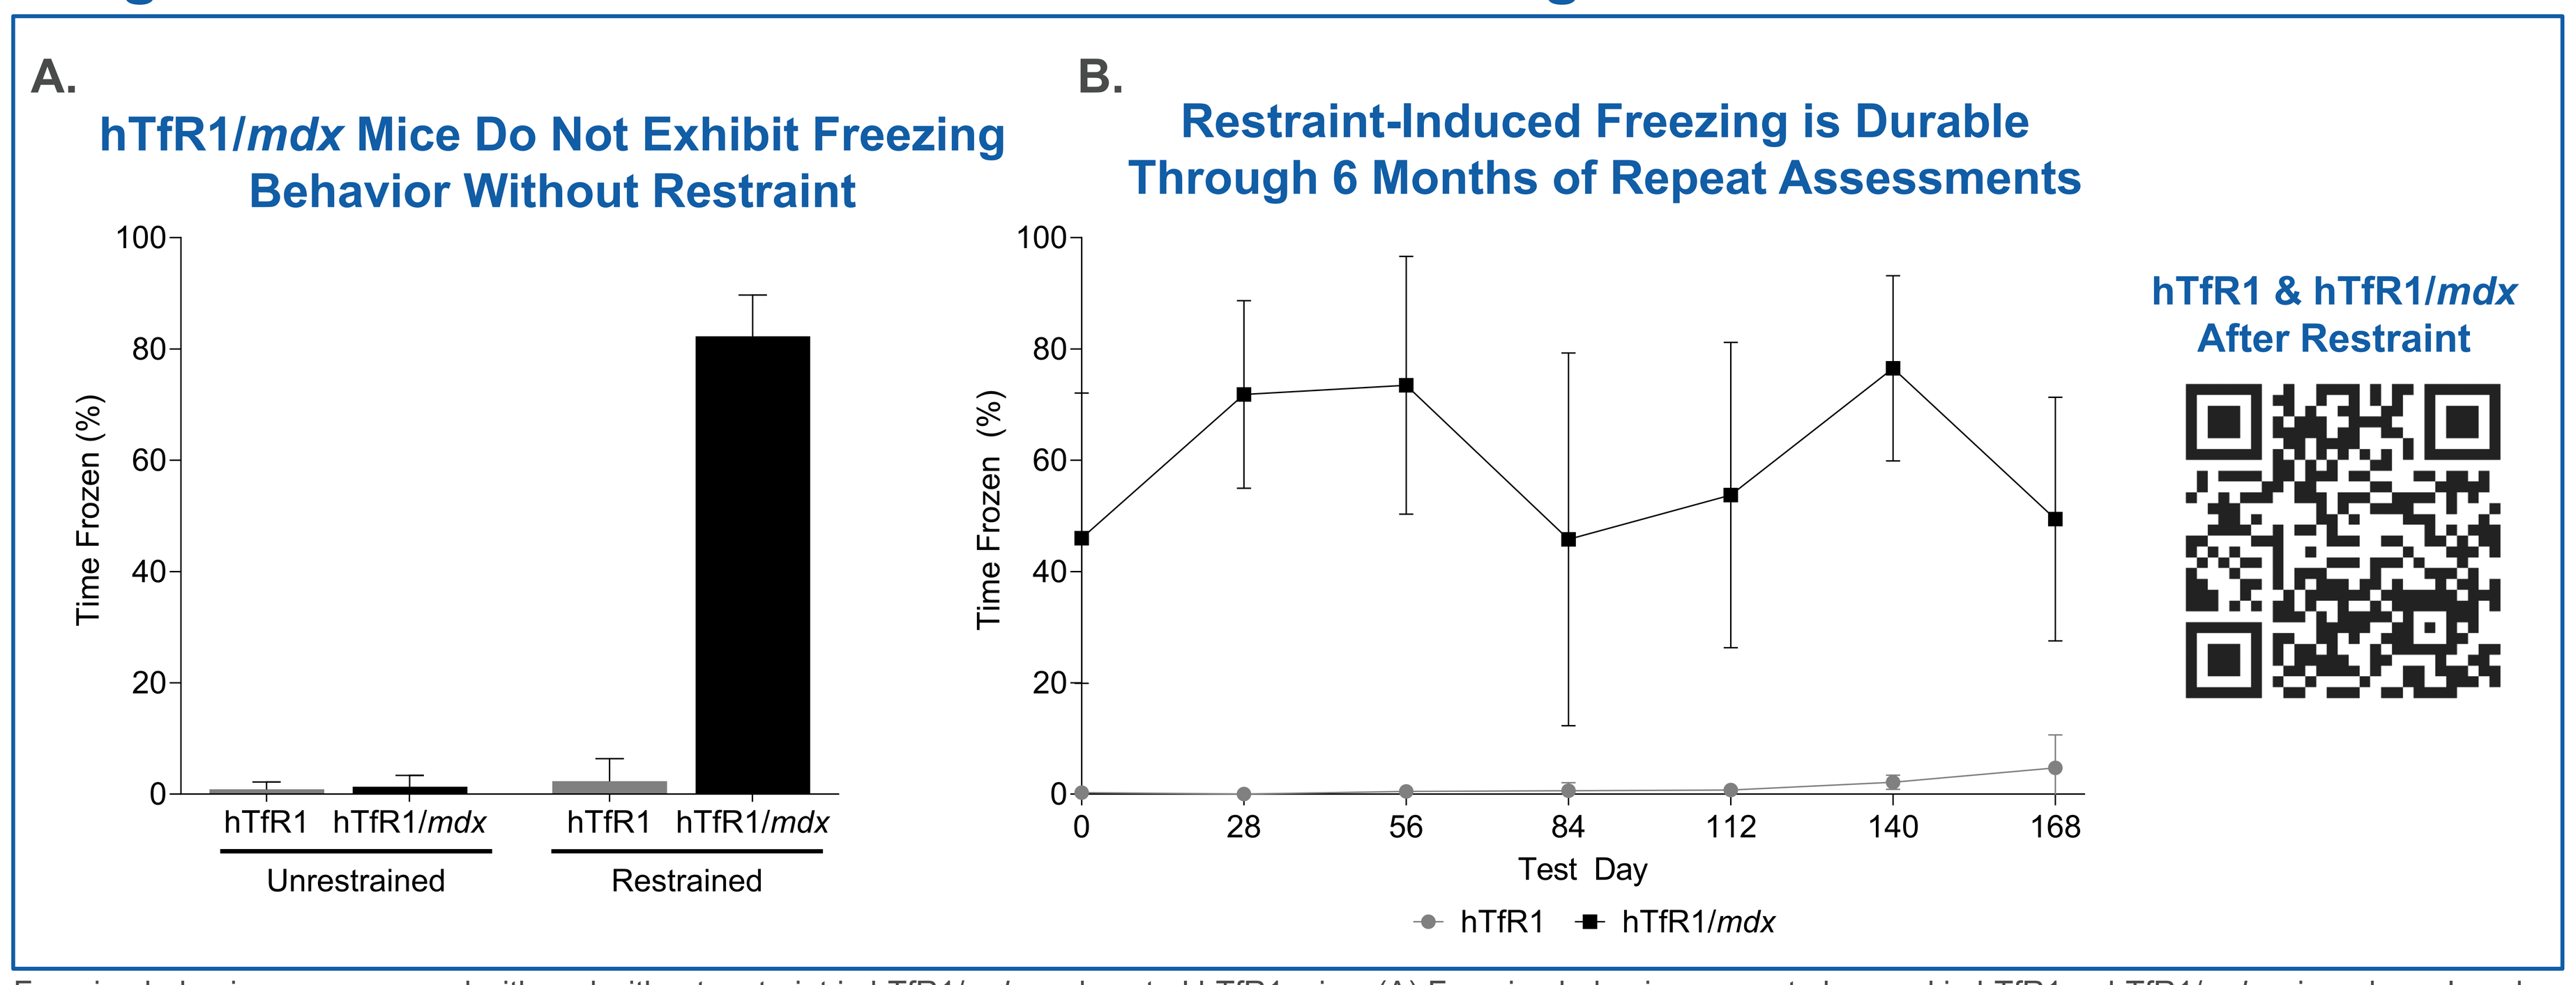
A.
hTfR1/mdx Mice Do Not Exhibit Freezing Behavior Without Restraint
Time Frozen (%)
hTfR1
hTfR1/mdx
Unrestrained
Restrained
B.
Restraint-Induced Freezing is Durable Through 6 Months of Repeat Assessments
Test Day
0
28
56
84
112
140
168
hTfR1 & hTfR1/mdx After Restraint

Freezing behavior was assessed with and without restraint in hTfR1/*mdx* and control hTfR1 mice. (A) Freezing behavior was not observed in hTfR1 or hTfR1/*mdx* mice when placed in the test cage without scruffing for ~15s. (B) Repeat longitudinal assessment of restraint-induced freezing showed durable behavioral phenotype in hTfR1/*mdx* mice and no freezing response in control hTfR1 mice. Freezing assessment was performed as outlined in Figure 3. Video clips were captured during D84 freezing assessment. Data are mean ±SD, n= 5 mice/group.

Figure 7. A Single Dose of FORCE-M23D Abolishes Restraint-Induced Freezing in hTfR1/*mdx* Mice Up To 2 Months Post-Dose

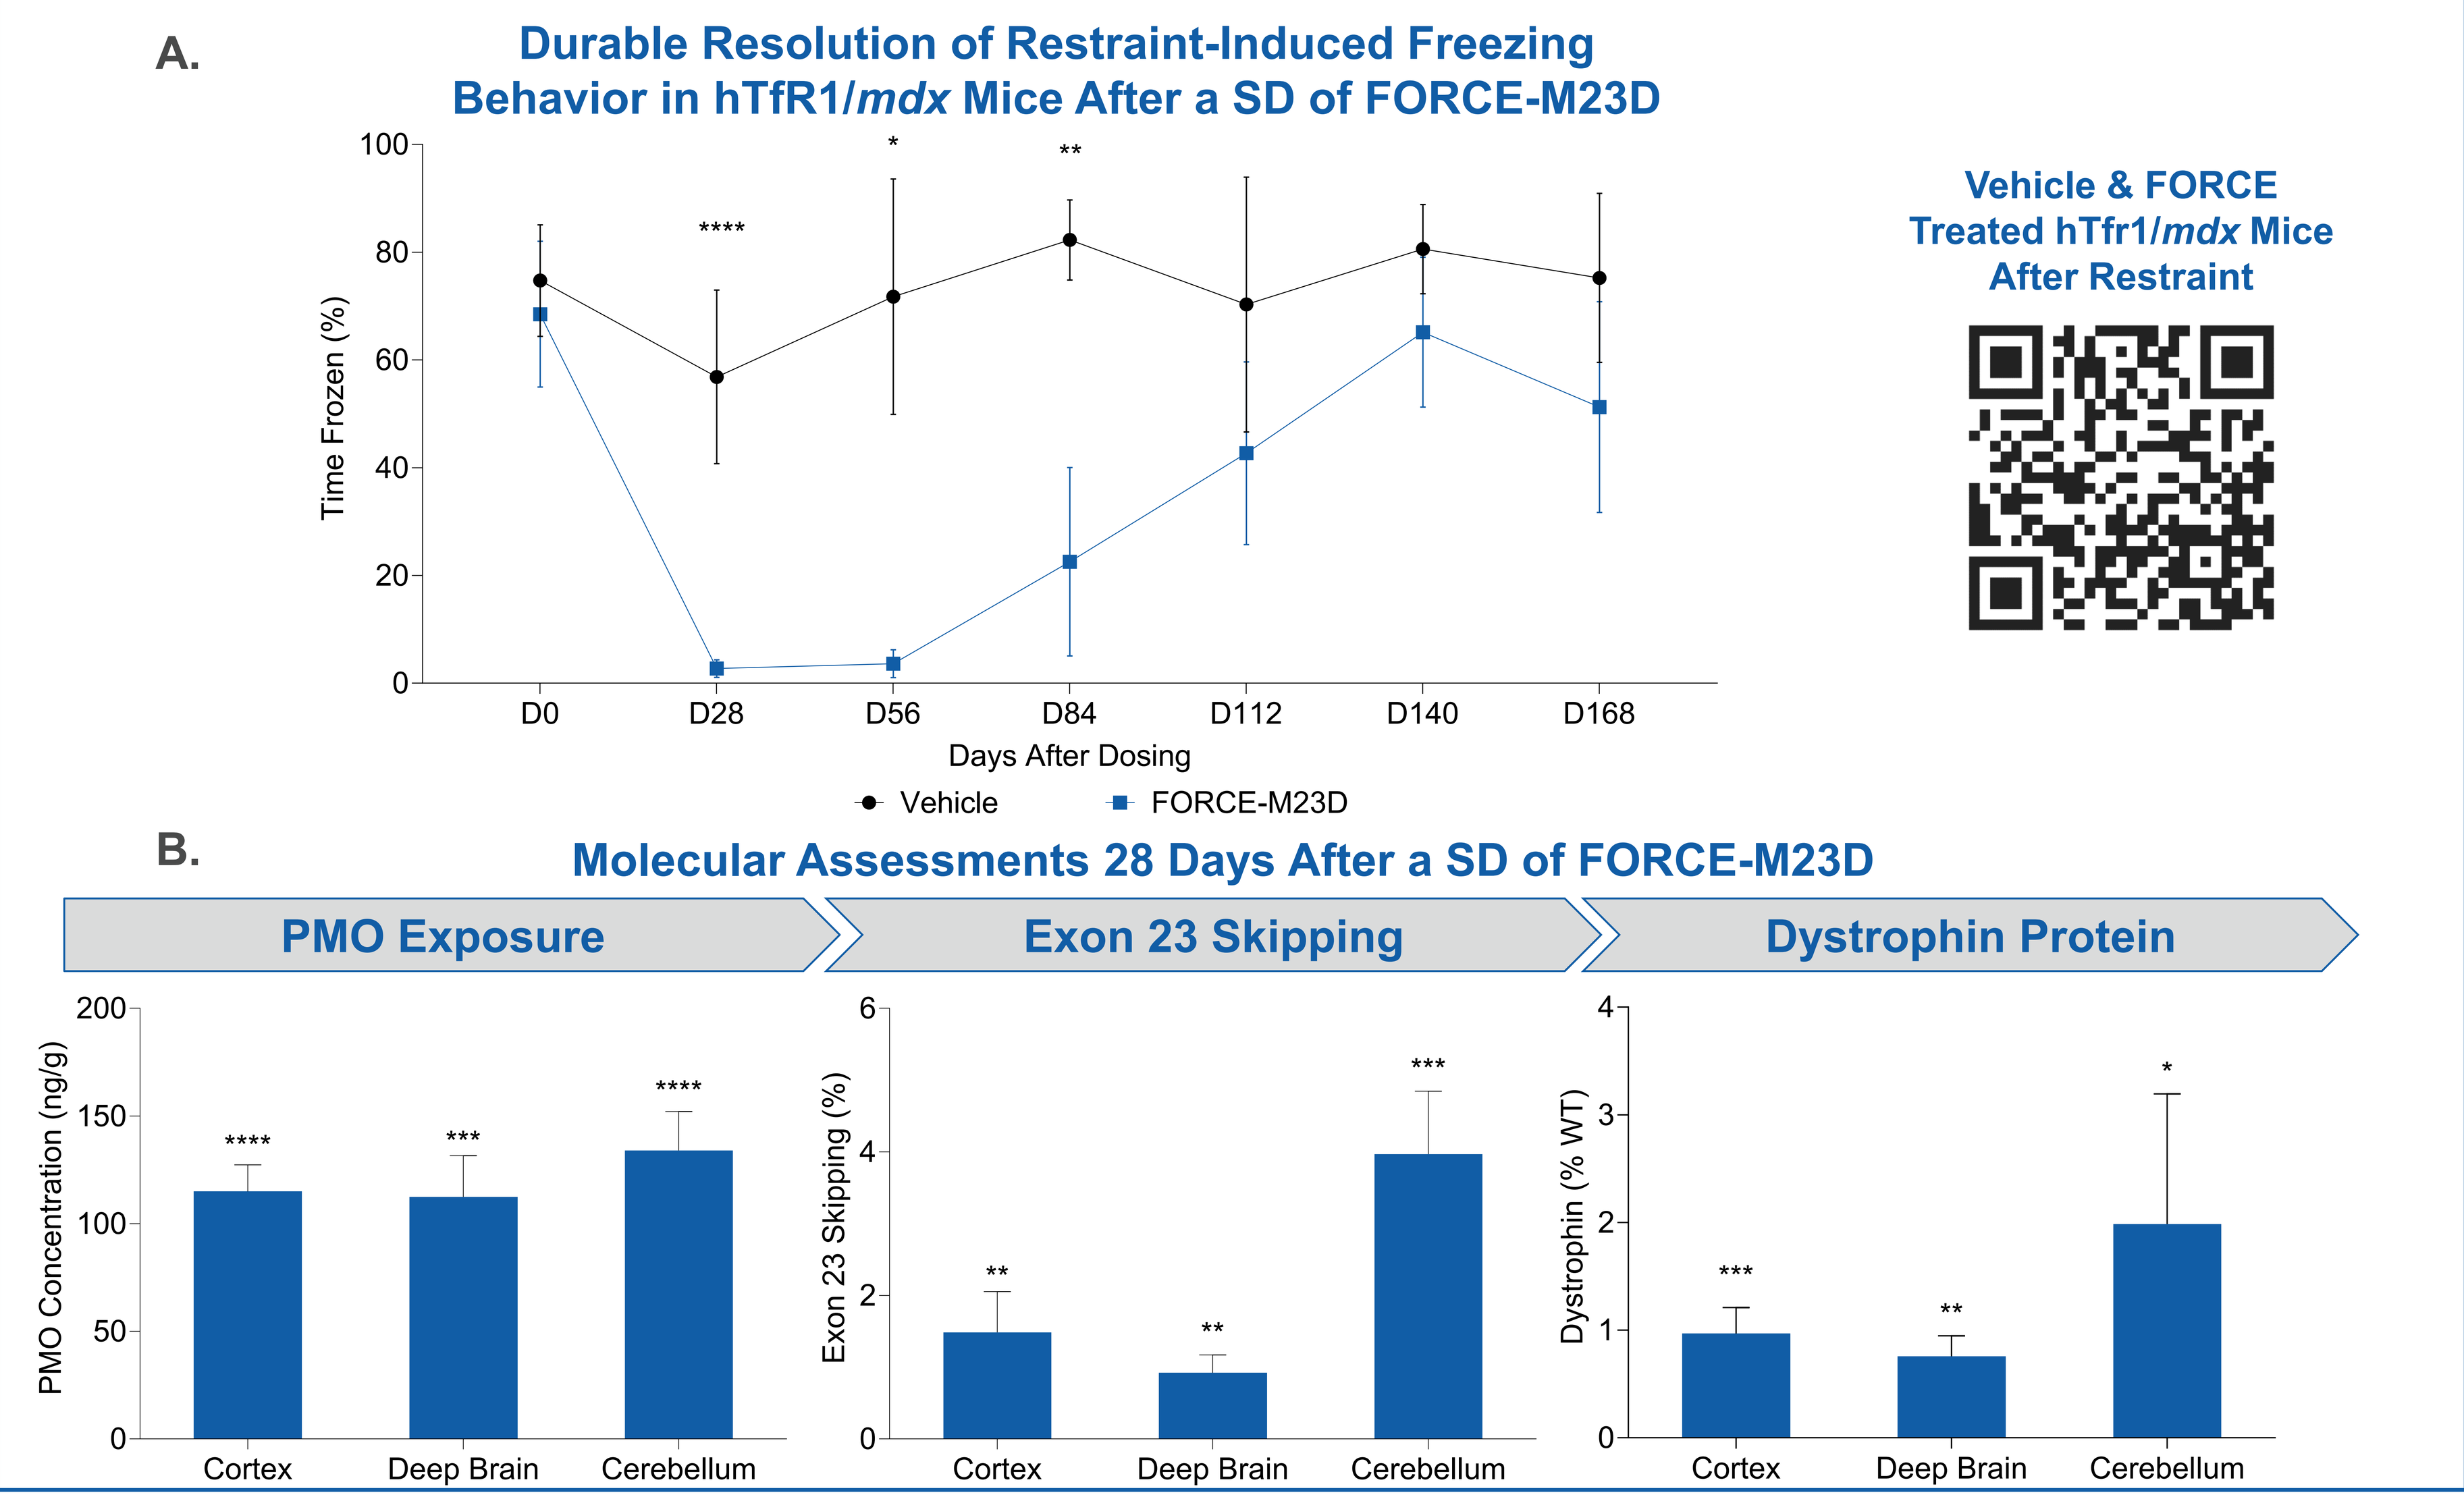
A.
Durable Resolution of Restraint-Induced Freezing Behavior in hTfR1/mdx Mice After a SD of FORCE-M23D
Time Frozen (%)
D0
D28
D56
D84
D112
D140
D168
Days After Dosing
Vehicle
FORCE-M23D
Vehicle & FORCE Treated hTfr1/mdx Mice After Restraint
B.
Molecular Assessments 28 Days After a SD of FORCE-M23D
PMO Exposure
Exon 23 Skipping
Dystrophin Protein
PMO Concentration (ng/g)
Exon 23 Skipping (%)
Dystrophin (% WT)
Cortex
Deep Brain
Cerebellum

A single dose of FORCE-M23D resolves restraint-induced freezing behavior in hTfR1/*mdx* mice up to 2 months post-dose. (A) Restraint-induced freezing was assessed at baseline and monthly after a single tail vein IV dose of vehicle or 30 mg/kg PMO equivalent FORCE-M23D for six months. Video clips were captured during D28 freezing assessment. Data are mean ±SD, n=10 mice/group at D0 & D28; n= 5 mice/group at all other timepoints; statistical analysis is two-way ANOVA test paired by timepoint with Sidak correction for multiple comparison. (B) Molecular assessment of cortex, deep brain, and cerebellum confirms broad PMO delivery (left), induction of exon 23 skipping (center), and restoration of dystrophin protein (right) 28 days after a single tail vein IV administration of FORCE-M23D. Data are mean ±SD, n=5 mice/group at D28; statistical analysis is unpaired t-test compared to vehicle treated hTfR1-*mdx* mice. * p<0.05; ** p<0.01; *** p<0.001; **** p<0.0001

CONCLUSIONS

- FORCE enables effective delivery of PMO to the CNS in the hTfR1/*mdx* model of DMD
- A single dose of FORCE-M23D leads to durable resolution of anxiety in hTfR1/*mdx* mice
- FORCE has the potential to address neurological manifestations of DMD

DISCLOSURE INFORMATION

All authors are former or current employees or advisors of Dyne Therapeutics, Inc. and may hold Dyne stock and/or stock options.

ACKNOWLEDGEMENTS

We would like to acknowledge the effort of Stephanie Rinaldi in supporting animal care & in-life portions of these studies.

REFERENCES

1. Anderson JL, Head SI, Rae C, Morley JW. Brain function in Duchenne muscular dystrophy. Brain. 2002;125(Pt 1):4-13.
2. Naidoo M, Anthony K. Dystrophin Dp71 and the Neuropathophysiology of Duchenne Muscular Dystrophy. Mol Neurobiol. 2020;57(3):1748-1767.
3. Saoudi A, Fergus C, Gileadi T, et al. Investigating the Impact of Delivery Routes for Exon Skipping Therapies in the CNS of DMD Mouse Models. Cells. 2023;12(6):908. Published 2023 Mar 15.
4. Zanotti S, Correia S, et al. The FORCE™ platform delivers oligonucleotides to the brain in the DM1 mouse model and in NHPs. ASGCT 2023
5. Desjardins CA, Yao M, Hall J, et al. Enhanced exon skipping and prolonged dystrophin restoration achieved by TfR1-targeted delivery of antisense oligonucleotide using FORCE conjugation in mdx mice. Nucleic Acids Res. 2022;50(20):11401-11414.
6. Sekiguchi M, Zushida K, Yoshida M, et al. A deficit of brain dystrophin impairs specific amygdala GABAergic transmission and enhances defensive behaviour in mice. Brain. 2009;132(Pt 1):124-135.

2025 UK Neuromuscular Translational Research Conference