221P

Initial Data From the ACHIEVE Trial of DYNE-101 in Adults With Myotonic Dystrophy Type 1 (DM1)

Daniel Wolf[1], James B. Lilleker[2], Guillaume Bassez[3], Jordi Diaz-Manera[4], Joost Kools[5], Marika Pane[6], Richard H Roxburgh[7], Benedikt Schoser[8], Christopher Turner[9], Chris Mix[1], Soma Ray[1], Baoguang Han[1], Wildon Farwell[1], Valeria Sansone[10]

[1]Dyne Therapeutics, Inc., Waltham, MA, USA; [2]Muscle Disease Unit, Northern Care Alliance NHS Foundation Trust, Manchester Academic Health Science Centre, Manchester, UK; [3]Institut de Myologie, Paris, France; [4]John Walton Muscular Dystrophy Research Centre, Newcastle University, Newcastle-Upon-Tyne, UK; [5]Radboud University Medical Center, Nijmegen, The Netherlands; [6]Fondazione Policlinico Universitario A. Gemelli, Rome, Italy; [7]Neurogenetics Clinic Centre for Brain Research, University of Auckland, Auckland, New Zealand; [8]Friedrich-Baur-Institute, Dep. of Neurology LMU Clinics, Ludwig-Maximilians University, Munich, Germany; [9]University College London Hospitals, London, UK; [10]Centro Clinico NEMO, University of Milan, Milan, Italy.

BACKGROUND

- Myotonic dystrophy type 1 (DM1) is a rare neuromuscular disorder with multisystem presentation. It is caused by expansion of CTG repeats in the 3′ untranslated region (3′UTR) of the dystrophia myotonica protein kinase (*DMPK*) gene. mRNA transcribed from the mutated gene forms hairpin-loop structures that sequester splicing regulators into toxic nuclear foci. This leads to widespread dysregulation of RNA splicing (spliceopathy) that drives the multisystem clinical manifestations.[1–3]
- No disease-modifying therapies are available, limiting treatment to symptom management.[4]
- DYNE-101, an investigational therapeutic for treatment of DM1, consists of a transferrin receptor 1 (TfR1)-binding antigen-binding fragment (Fab) conjugated to an antisense oligonucleotide (ASO) designed against mutant nuclear *DMPK* mRNA to correct splicing.[5,6]
- The safety and efficacy of DYNE-101 are being investigated in the Phase 1/2 ACHIEVE trial (NCT05481879; EudraCT number 2022-000889-18).[6,7]

METHODS

- ACHIEVE is a global, randomized, placebo-controlled study evaluating once monthly or less frequent intravenous administrations of DYNE-101 in adults (18–49 years) with DM1. It consists of a multiple ascending dose (MAD) period (24 weeks; **Figure 1**), an open-label extension (OLE) period (24 weeks), and a long-term extension (LTE) period (96 weeks).[6,7]
 - 56 participants have received DYNE-101 across five different dose/dose regimen cohorts in the MAD portion of the study (**Figure 1**). All patients will receive the highest tolerable dose of DYNE-101 during the OLE and LTE periods.[6]
 - In the MAD portion of the study, muscle biopsies are collected at baseline, 12 weeks, and 24 weeks.[6]
 - The primary endpoints are safety and tolerability.[6,7]
 - Additional endpoints include pharmacokinetics and pharmacodynamics, including change from baseline in splicing, multiple assessments of muscle strength and function, and patient-reported outcomes (PROs; including Myotonic Dystrophy type 1 Activity and participation scale [DM1-ACTIV[c]] and Myotonic Dystrophy Health Index [MDHI]).[6,7]

Figure 1. Design of the MAD Portion of the ACHIEVE Study

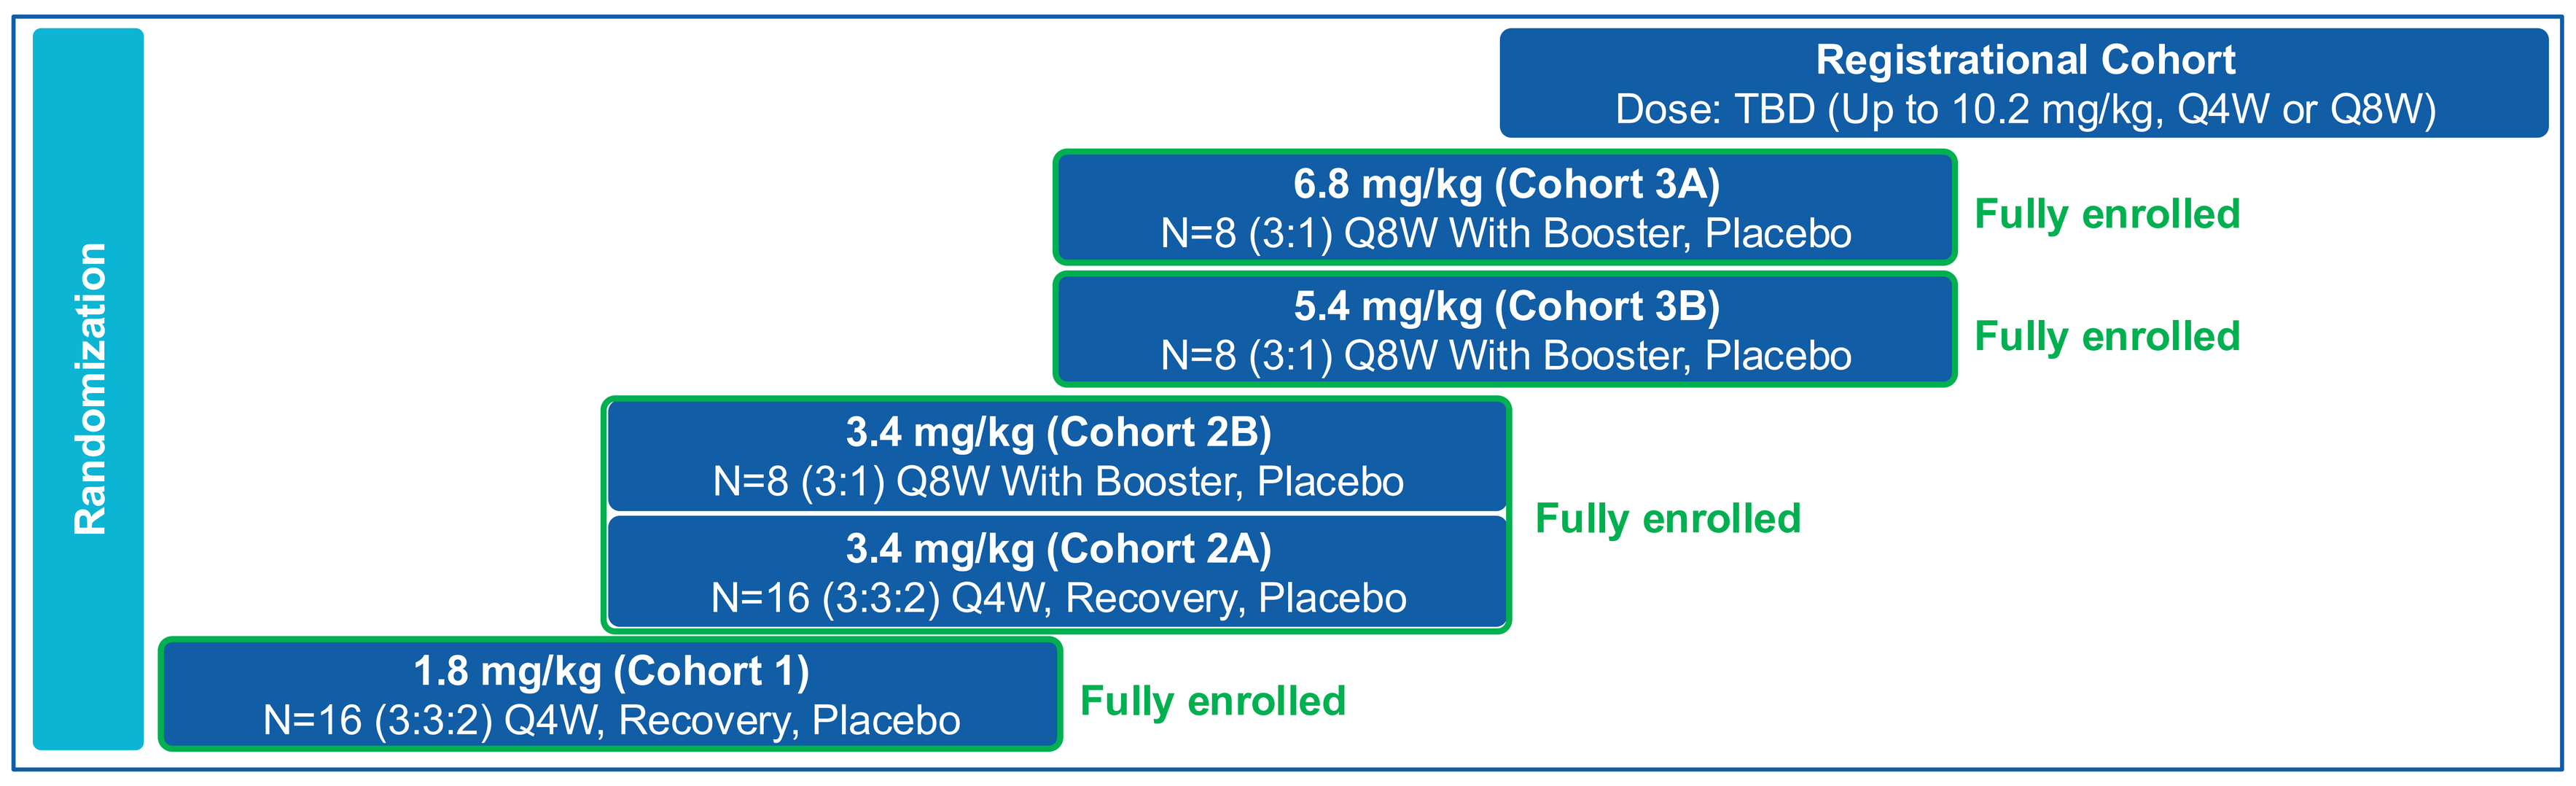

Randomization
Registrational Cohort
Dose: TBD (Up to 10.2 mg/kg, Q4W or Q8W)
6.8 mg/kg (Cohort 3A)
N=8 (3:1) Q8W With Booster, Placebo
Fully enrolled
5.4 mg/kg (Cohort 3B)
N=8 (3:1) Q8W With Booster, Placebo
Fully enrolled
3.4 mg/kg (Cohort 2B)
N=8 (3:1) Q8W With Booster, Placebo
3.4 mg/kg (Cohort 2A)
N=16 (3:3:2) Q4W, Recovery, Placebo
Fully enrolled
1.8 mg/kg (Cohort 1)
N=16 (3:3:2) Q4W, Recovery, Placebo
Fully enrolled

Doses provided refer to ASO component of DYNE-101. Recovery cohort will receive Q4W x 2 doses then placebo for the remainder of the 24-week placebo-controlled period. Q8W with booster includes Q4W x 3 doses then Q8W dosing. Study protocol allows for dosing up to 10.2 mg/kg.
ASO, antisense oligonucleotide; MAD, multiple ascending dose; Q4W, every 4 weeks; Q8W, every 8 weeks; TBD, to be determined.

- Here we present 12-month efficacy data from the 1.8 mg/kg every 4 weeks (Q4W) cohort, 6-month data from the 3.4 mg/kg Q4W cohort, and 3-month data from the 5.4 mg/kg every 8 weeks (Q8W) cohort. Safety data are as of August 20, 2024, and include all 56 participants dosed through 6.8 mg/kg Q8W.

RESULTS

Table 1. Baseline Characteristics

Mean (SD) or n (%)	1.8 mg/kg Q4W (N=16)[a]	3.4 mg/kg Q4W (N=16)[a]	5.4 mg/kg Q8W (N=8)[b]
Age (years)	34.6 (10.4)	34.3 (7.6)	39.6 (7.0)
Female, n (%)	7 (43.8%)	3 (18.8%)	5 (62.5%)
BMI (kg/m²)	22.4 (5.3)	23.8 (3.8)	21.7 (2.7)
CASI	0.62 (0.26)	0.67 (0.20)	0.79 (0.14)
CTG repeats	375 (217)	527 (241)	586 (294)
vHOT (sec) (middle finger)	11.2 (4.3)	8.0 (5.7)	10.1 (6.2)
QMT total (% predicted)	49.6 (10.9)	47.8 (10.6)	45.8 (16.1)
5 times sit to stand (sec)	9.33 (2.02)	10.05 (3.03)	12.28 (5.96)
DM1-ACTIV[c] total	43 (7)	42 (7)	44 (6)
MDHI total	25 (20)	25 (20)	16 (9)

a. Q4W, recovery, and placebo arms are reported together for baseline characteristics.
b. Q8W and placebo arms are reported together for baseline characteristics.
BMI, body mass index; CASI, composite alternative splicing index;
CTG, cytosine, thymine and guanine; DM1-ACTIV[c], Myotonic Dystrophy type 1 Activity and participation scale; Q4W, every 4 weeks; Q8W, every 8 weeks; QMT, quantitative muscle testing; MDHI, Myotonic Dystrophy Health Index; SD, standard deviation; sec, seconds; vHOT, video hand opening time.

Table 2. DYNE-101 Safety Profile Is Favorable to Date (Summary of TEAEs)

TEAE Category	Participants With ≥1 TEAE – n (%)					
	1.8 mg/kg Q4W+Rec. N=16	3.4 mg/kg Q4W+Rec. N=16	3.4 mg/kg Q8W N=8	5.4 mg/kg Q8W N=8	6.8 mg/kg Q8W N=8	Overall (N=56)
Any TEAE	16 (100%)	16 (100%)	8 (100%)	8 (100%)	7 (88%)	55 (98%)
Any related TEAE	8 (50%)	8 (50%)	2 (25%)	3 (38%)	5 (63%)	26 (46%)
Any serious TEAE	4 (25%)	0	1 (13%)	0	0	5 (9%)
Any serious related TEAE	0	0	0	0	0	0
Any TEAE leading to withdrawal	0	0	0	0	0	0
Any TEAE leading to death	0	0	0	0	0	0

Data as of August 20, 2024.
OLE, open-label extension; Q4W, every 4 weeks; Q8W, every 8 weeks; Rec, recovery; TEAE, treatment-emergent adverse events.

- 6 serious treatment-emergent adverse events (TEAEs) unrelated to study drug
 - Atrioventricular block first degree (1)[a]
 - Pneumonia (2 events in same participant)
 - Pulmonary embolism (1)[b]
 - Hyponatremia (1)
 - Influenza (1)
- Most common TEAEs (≥20% participant incidence)[c]
 - Nasopharyngitis (32%)
 - Procedural pain (29%)
 - Infusion-related reaction (21%)
- Liver enzyme elevations have been observed in a minority of participants
 - No impact on liver function (bilirubin or coagulation)
 - Interpretation is complicated by underlying disease and elevated baseline values up to ~2.5x greater than the upper limit of normal
- No participants have demonstrated persistent related anemia or thrombocytopenia

a. Transient worsening of atrioventricular (AV) block in a participant with ongoing medical history of first-degree AV block. b. Attributed to risk factors for pulmonary embolism. c. All cohorts combined; preferred terms are reported.

Figure 2. Monthly Dosing of DYNE-101 Demonstrated Dose-Dependent ASO Delivery at 3 Months

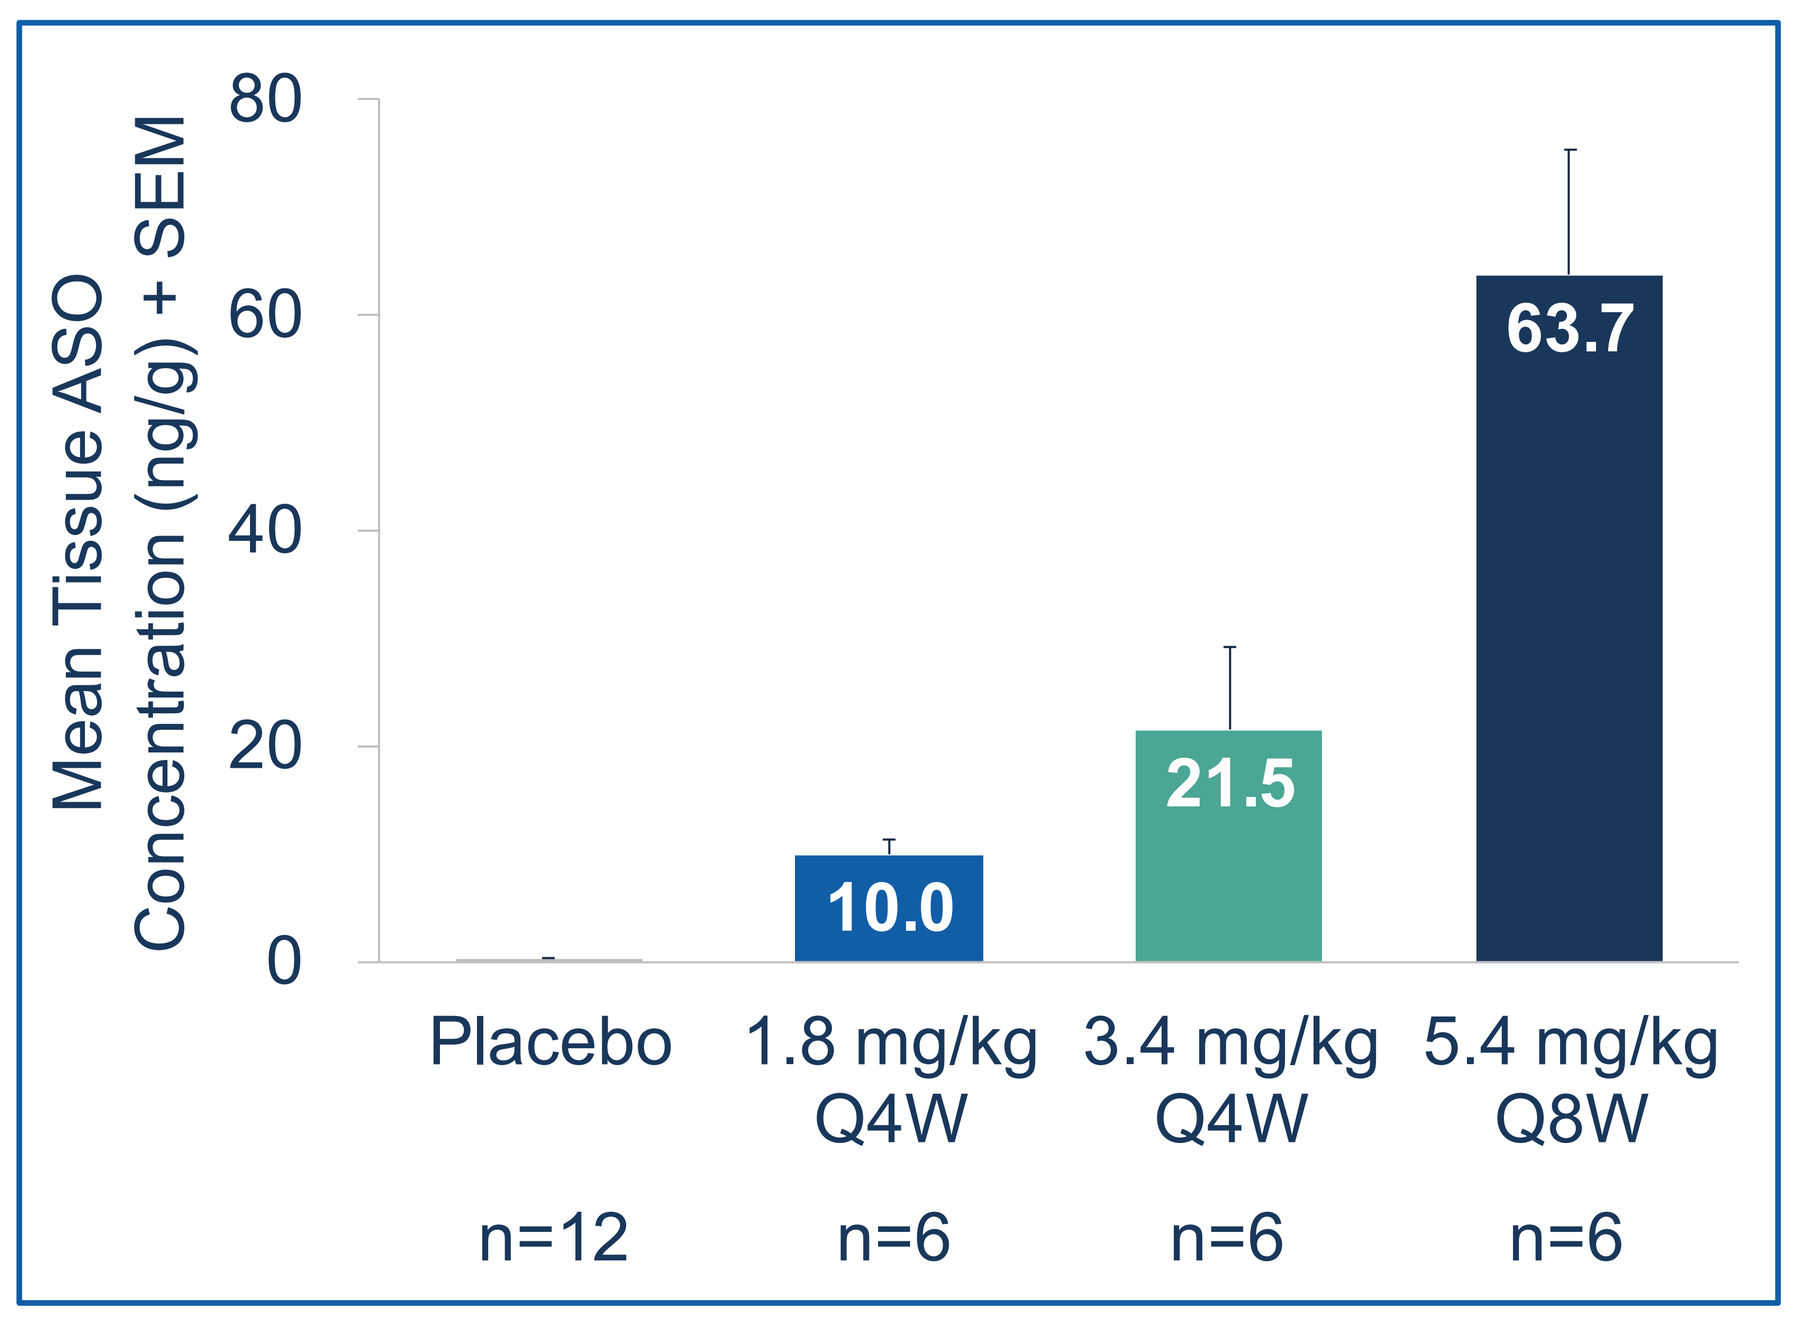

Mean Tissue ASO Concentration (ng/g) + SEM
80
60
40
20
0
10.0
21.5
63.7
Placebo
1.8 mg/kg Q4W
3.4 mg/kg Q4W
5.4 mg/kg Q8W
n=12
n=6
n=6
n=6

ASO, antisense oligonucleotide; Q4W, every 4 weeks; Q8W, every 8 weeks; SEM, standard error of mean.

Figure 3. Monthly Dosing of DYNE-101 Demonstrated Consistent Splicing Correction at 3 Months (CASI-22)[a]

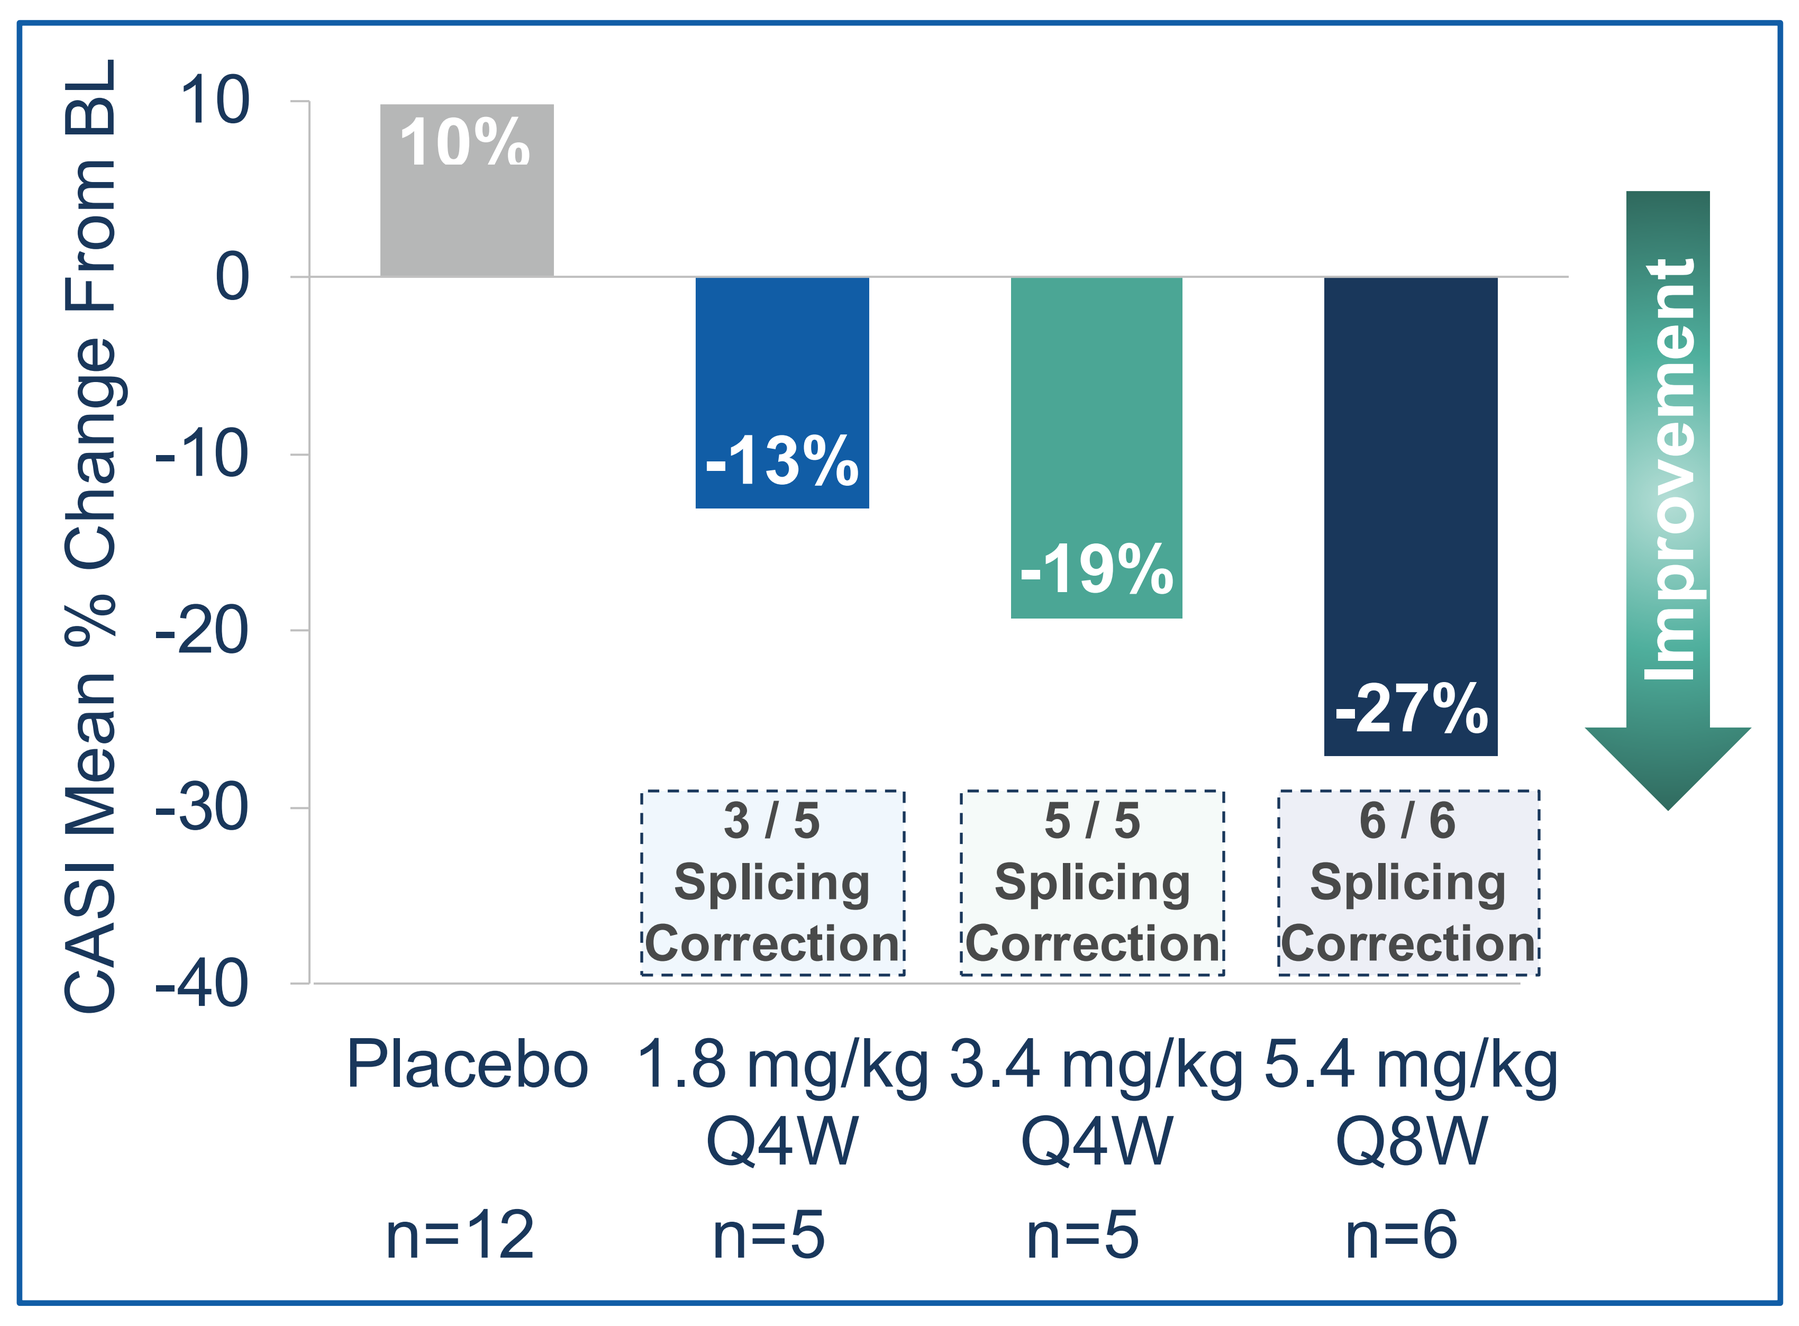

CASI Mean % Change From BL
10
0
-10
-20
-30
-40
10%
-13%
-19%
-27%
3 / 5 Splicing Correction
5 / 5 Splicing Correction
6 / 6 Splicing Correction
Improvement
Placebo
1.8 mg/kg Q4W
3.4 mg/kg Q4W
5.4 mg/kg Q8W
n=12
n=5
n=5
n=6

a. CASI-22 is a validated composite alternative splicing index (CASI) comprising 22 genes implicated in pathways that cause the symptoms of DM1.[8,9]
One post-baseline sample in the 3.4 mg/kg Q4W treatment group not included within splicing assay as the sample did not meet quality control criteria.
BL, baseline; CASI, composite alternative splicing index; DM1, myotonic dystrophy type 1; Q4W, every 4 weeks; Q8W, every 8 weeks.

Figure 4. DYNE-101 Demonstrated Improvements in Functional Endpoints, Including Measures of Myotonia and Muscle Strength

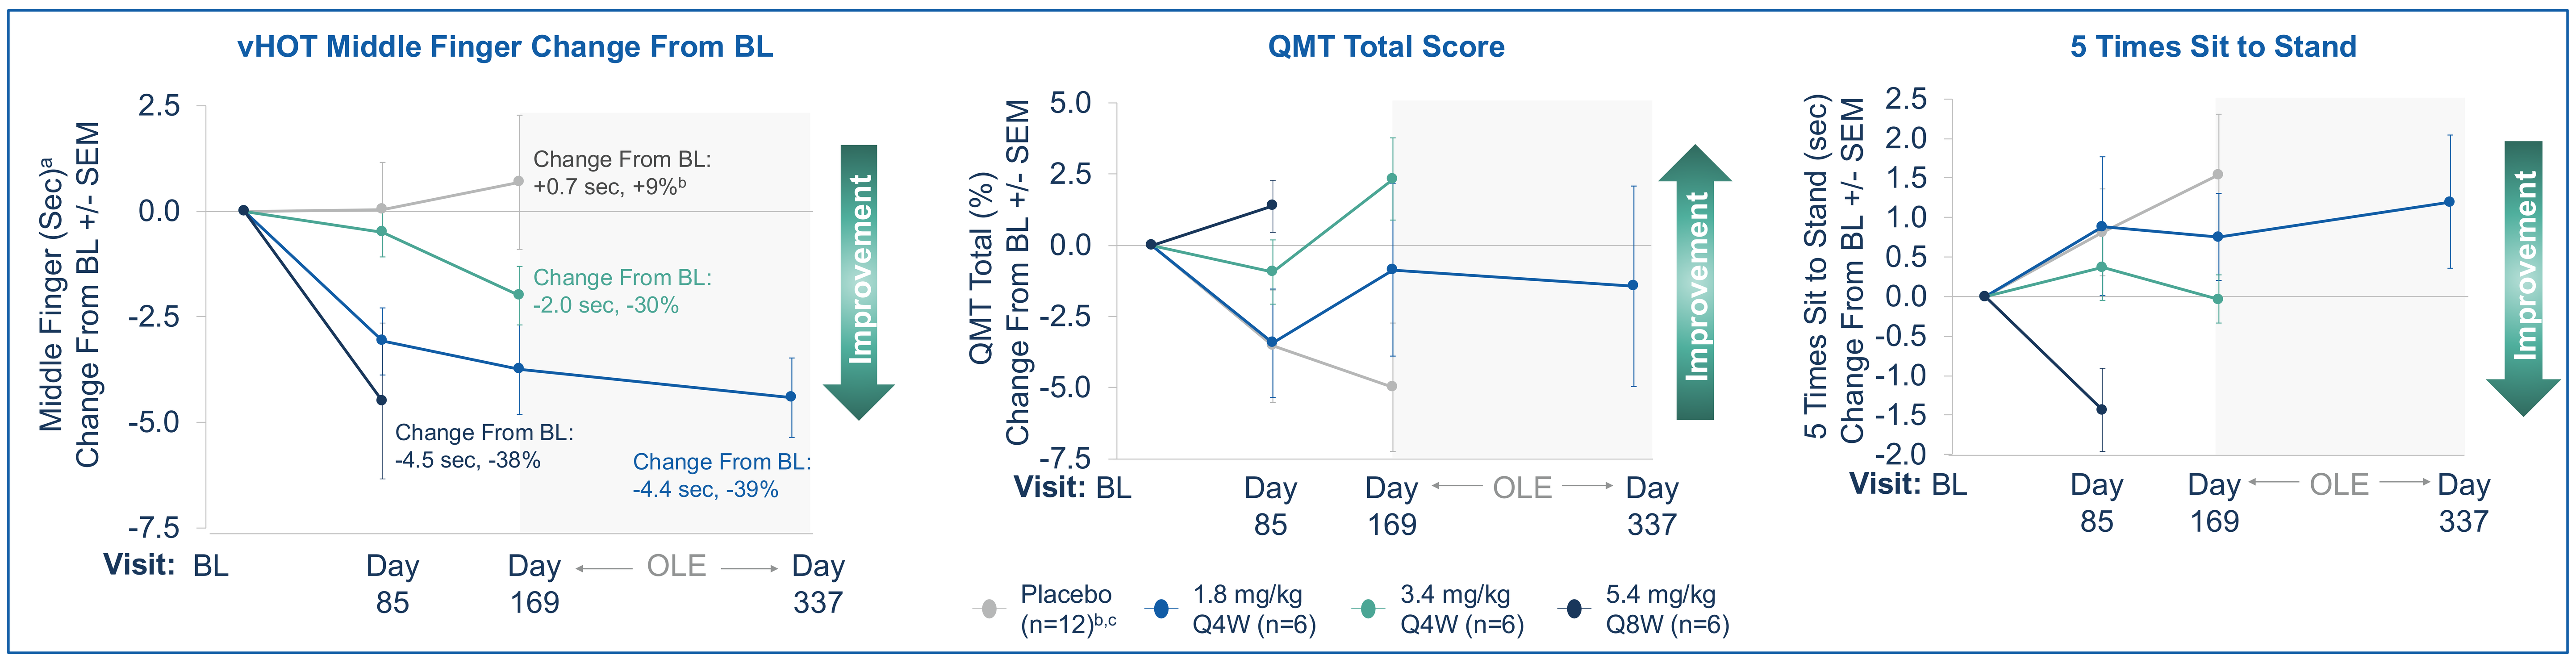

vHOT Middle Finger Change From BL
Middle Finger (Sec)[a] Change From BL +/- SEM
2.5
0.0
-2.5
-5.0
-7.5
Change From BL: +0.7 sec, +9%[b]
Change From BL: -2.0 sec, -30%
Change From BL: -4.5 sec, -38%
Change From BL: -4.4 sec, -39%
Improvement
Visit: BL
Day 85
Day 169
OLE
Day 337
QMT Total Score
QMT Total (%) Change From BL +/- SEM
5.0
2.5
0.0
-2.5
-5.0
-7.5
Improvement
Visit: BL
Day 85
Day 169
OLE
Day 337
5 Times Sit to Stand
5 Times Sit to Stand (sec) Change From BL +/- SEM
2.5
2.0
1.5
1.0
0.5
0.0
-0.5
-1.0
-1.5
-2.0
Improvement
Visit: BL
Day 85
Day 169
OLE
Day 337
Placebo (n=12)[b,c]
1.8 mg/kg Q4W (n=6)
3.4 mg/kg Q4W (n=6)
5.4 mg/kg Q8W (n=6)

a. Middle Finger (sec) is the average of all myotonia trials for an individual participant in ACHIEVE. b. Mean percent change from baseline for placebo group is based on baseline values from 12 patients. c. Placebo group includes 12 participants at Day 85 and 8 participants at Day 169.
BL, baseline; OLE, open-label extension; Q4W, every 4 weeks; Q8W, every 8 weeks; QMT, quantitative muscle testing; sec, seconds; SEM, standard error of mean; vHOT, video hand opening time.

Figure 5. DYNE-101 Showed Improvement From Baseline in Activities of Daily Living (DM1-ACTIV[c])

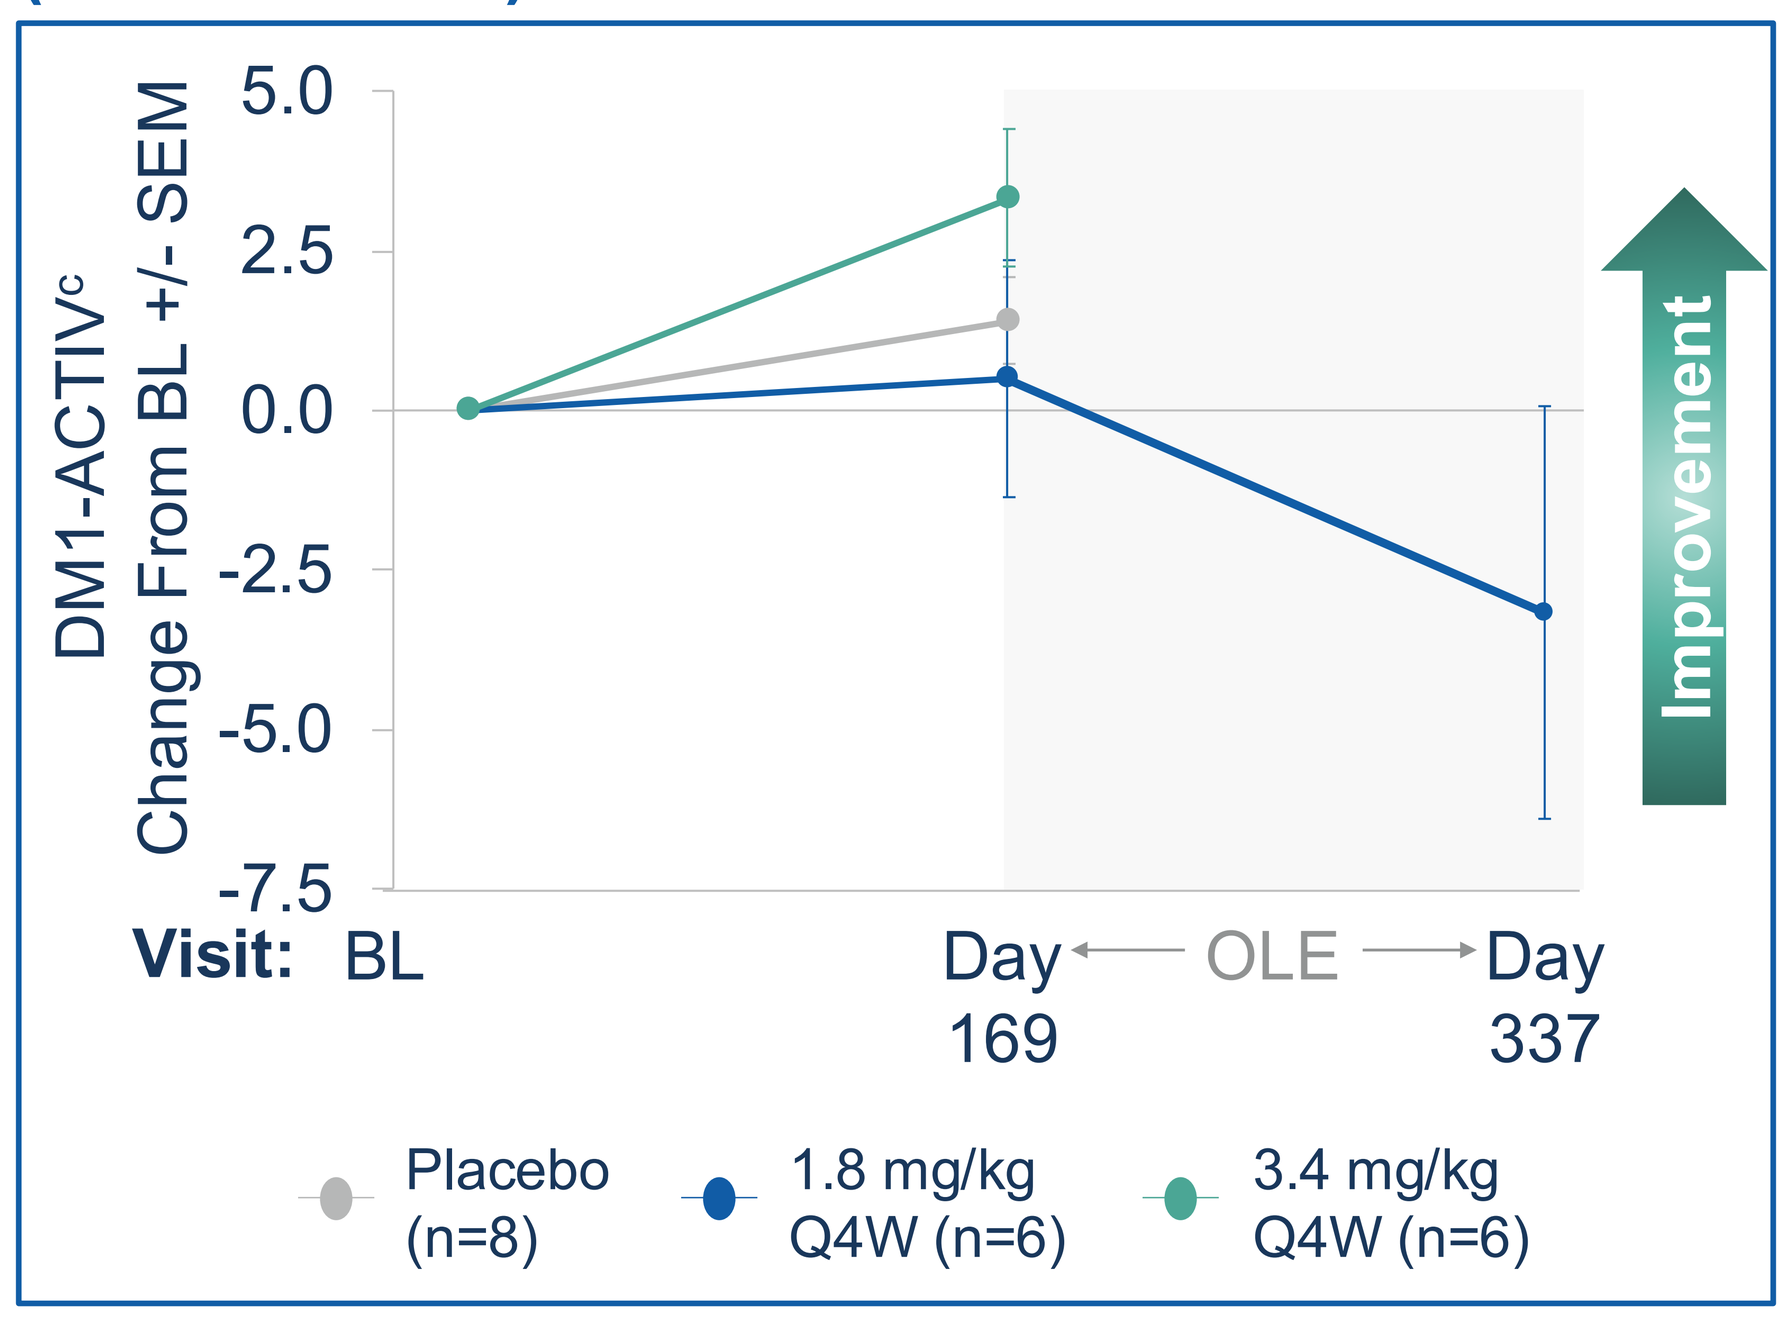

DM1-ACTIV[c] Change From BL +/- SEM
5.0
2.5
0.0
-2.5
-5.0
-7.5
Improvement
Visit: BL
Day 169
OLE
Day 337
Placebo (n=8)
1.8 mg/kg Q4W (n=6)
3.4 mg/kg Q4W (n=6)

PROs collected at BL, Day 169, and Day 337.
BL, baseline; DM1-ACTIV[c], Myotonic Dystrophy type 1 Activity and participation scale; OLE, open-label extension; PRO, patient-reported outcomes; Q4W, every 4 weeks; SEM, standard error of mean.

Figure 6. DYNE-101 Demonstrated Clinical Benefit Based on Well-Validated PRO (MDHI Total Score)

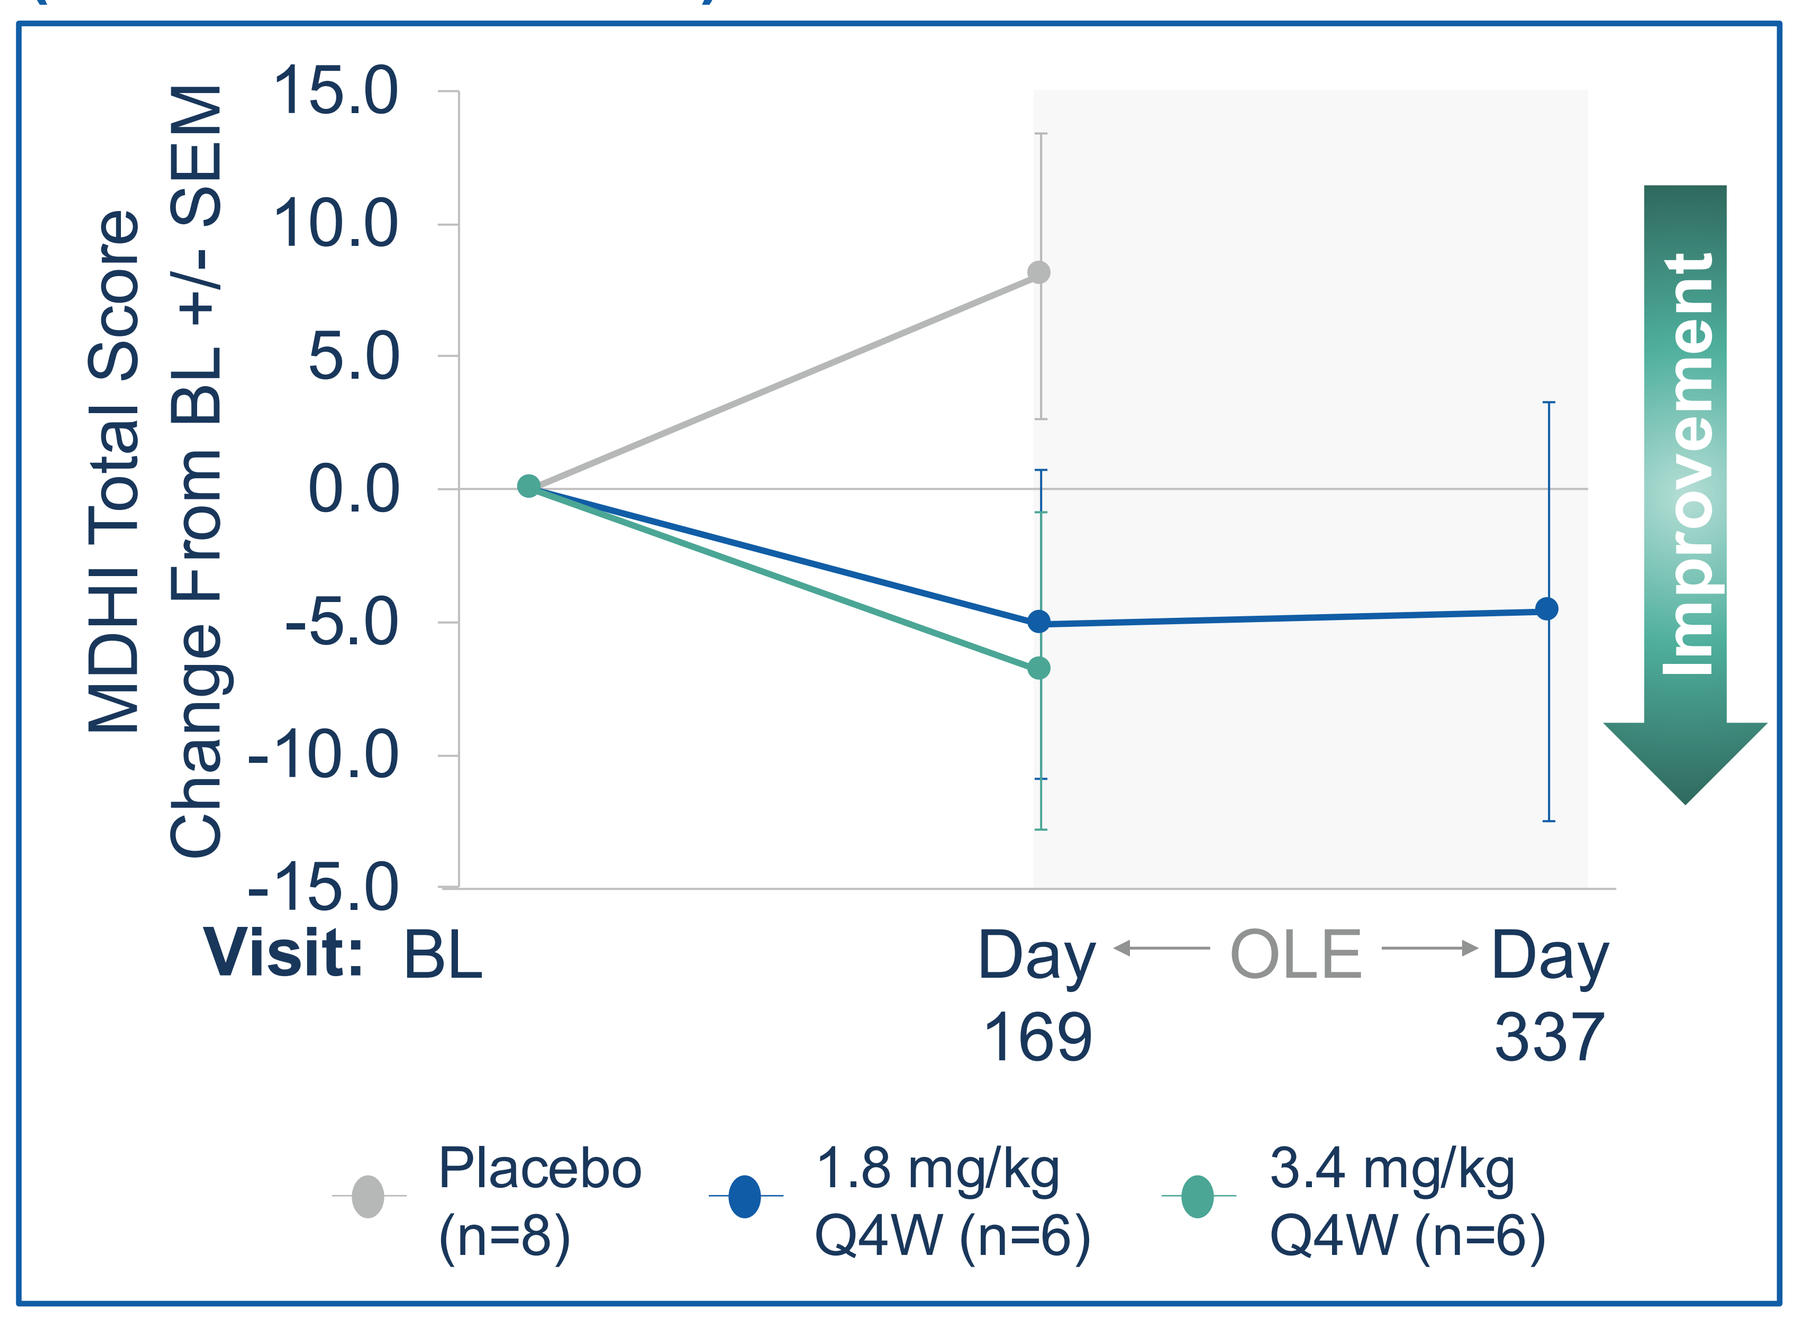

MDHI Total Score Change From BL +/- SEM
15.0
10.0
5.0
0.0
-5.0
-10.0
-15.0
Improvement
Visit: BL
Day 169
OLE
Day 337
Placebo (n=8)
1.8 mg/kg Q4W (n=6)
3.4 mg/kg Q4W (n=6)

PROs collected at BL, Day 169, and Day 337.
BL, baseline; MDHI, Myotonic Dystrophy Health Index; OLE, open-label extension; PRO, patient-reported outcomes; Q4W, every 4 weeks; SEM, standard error of mean.

CONCLUSIONS

- DYNE-101 has demonstrated a favorable safety profile to date.[a]
- DYNE-101 demonstrated dose-dependent muscle delivery and consistent splicing correction.
- DYNE-101 led to meaningful improvements in multiple clinical endpoints, including myotonia, muscle strength, timed functional assessments, and PROs.
- The data support the continued development of DYNE-101 for the treatment of DM1.

a. Data as of August 20, 2024.

REFERENCES

1. Lopez-Martinez A, et al. *Genes (Basel).* 2020;11:1109;
2. Pavićević DS, et al. *Biomed Res Int.* 2013;2013:391821;
3. Thornton CA. *Neurol Clin.* 2014;32(3):705–719;
4. Thornton CA, et al. *Curr Opin Genet Dev.* 2017;44:135–140;
5. Zanotti S, et al. Poster presented at: American Society of Gene and Cell Therapy Annual Meeting; May 16–20, 2023; Washington, USA;
6. Dyne Corporate presentation. September 2024;
7. ClinicalTrials.gov. NCT05481879 (https://clinicaltrials.gov/ct2/show/NCT05481879);
8. Wang W. 2017. University of Rochester School of Medicine and Dentistry PhD thesis. Available at: http://hdl.handle.net/1802/32572 (accessed September 5, 2024);
9. Provenzano M, et al. bioRxiv 2024. Available at: https://www.biorxiv.org/content/10.1101/2024.07.10.602610v1 (accessed September 5, 2024).

DISCLOSURE INFORMATION

Daniel Wolf, Chris Mix, Soma Ray, Baoguang Han, and Wildon Farwell are employees of Dyne Therapeutics and may hold stock in the company; James B. Lilleker has participated in advisory boards and/or conference support/presentations for Roche, Sanofi, and Dyne Therapeutics; Jordi Diaz-Manera has received funding for participating on advisory boards or presenting at conferences on behalf of Sanofi, Sarepta, Lupin, Amicus, and Astellas. He has received funding for research from Sanofi, Sarepta, Spark and Boehringer-Ingelheim; Joost Kools has nothing to disclose; Marika Pane has nothing to disclose; Richard H Roxburgh has nothing to disclose; Benedikt Schoser has received unrestricted research grants from Amicus, Astellas, Roche, Marigold Foundation, AMDA Foundation, and speaker's honoraria from Amicus Therapeutics Inc., Alexion, Kedrion, and Sanofi. He has participated as a scientific advisor for Amicus Therapeutics Inc., Argenx, Astellas, Bayer, Pepgen, Sanofi, Spark, and Taysha. He declares no stocks or shares; Christopher Turner has acted as a consultant for PepGen and Vertex Pharmaceuticals; Valeria Sansone has received compensation for intellectual and teaching activities from Dyne Therapeutics, Avidity Biosciences, Roche, Biogen, Novartis, and Lupin Pharmaceuticals.

ACKNOWLEDGMENTS

This study was funded by Dyne Therapeutics. Medical writing and editorial support was provided by Olivia Atkinson of Obsidian Healthcare Group Ltd, UK. This support was funded by Dyne Therapeutics. The authors wish to thank the clinical trial investigators and site coordinators and, most importantly, all the patients, families, and caregivers for their willingness to participate in this clinical trial, which was sponsored by Dyne Therapeutics.

World Muscle Society Annual Congress, Prague, Czechia, October 8–12, 2024