669P

The FORCE™ Platform Enables TfR1-mediated Delivery of Enzyme Replacement Therapy to Muscle and Central Nervous System, Resolving Pompe Pathology in Mice

Tyler Picariello, Benjamin Vieira, Jennifer Johnson, Lydia Schlaefke, Ann (Ya-An) Chang, Jin Cui, Sihyung Yang, Stephanie Rinaldi, Timothy Weeden, Stefano Zanotti, Oxana Beskrovnaya

Dyne Therapeutics Inc., Waltham, MA, USA

BACKGROUND

Pompe disease is a severe neuromuscular disorder caused by deficiency of the lysosomal enzyme acid alpha glucosidase (GAA). Lack of GAA leads to glycogen accumulation in muscle and subsequent weakness, cardiomyopathy, and respiratory failure[1-5]. Enzyme replacement therapy (ERT) with recombinant human GAA is the standard of care (SOC) and increases survival but has inadequate efficacy in skeletal muscle. Importantly, Pompe is also characterized by CNS manifestations, including behavioral and cognitive deficits due to glycogen accumulation in CNS cells, which are not addressed by the SOC[6-8].

The FORCE platform enables TfR1-mediated delivery of therapeutics via a TfR1-targeting fragment antigen binding (Fab). We engineered the platform with GAA as payload (FORCE-GAA, Fig.1) to enhance GAA uptake in muscle and enable CNS delivery. To evaluate FORCE-GAA efficacy *in vivo*, we crossed mice expressing human TfR1 with 6^{Neo} mice[9], a well-established model of Pompe, to create hTfR1/6^{Neo} mice. Intravenous injections of FORCE-GAA in hTfR1/6^{Neo} mice cleared glycogen in muscle after 8 weeks and normalized lysosomal size, confirming restoration of GAA activity in the lysosome. FORCE-GAA displayed superior efficacy and dose potency relative to GAA. These data demonstrate the potential of FORCE-GAA as a novel therapy for Pompe and validate FORCE as a versatile drug delivery platform.

Figure 1. FORCE-GAA Enables TfR1-targeted ERT

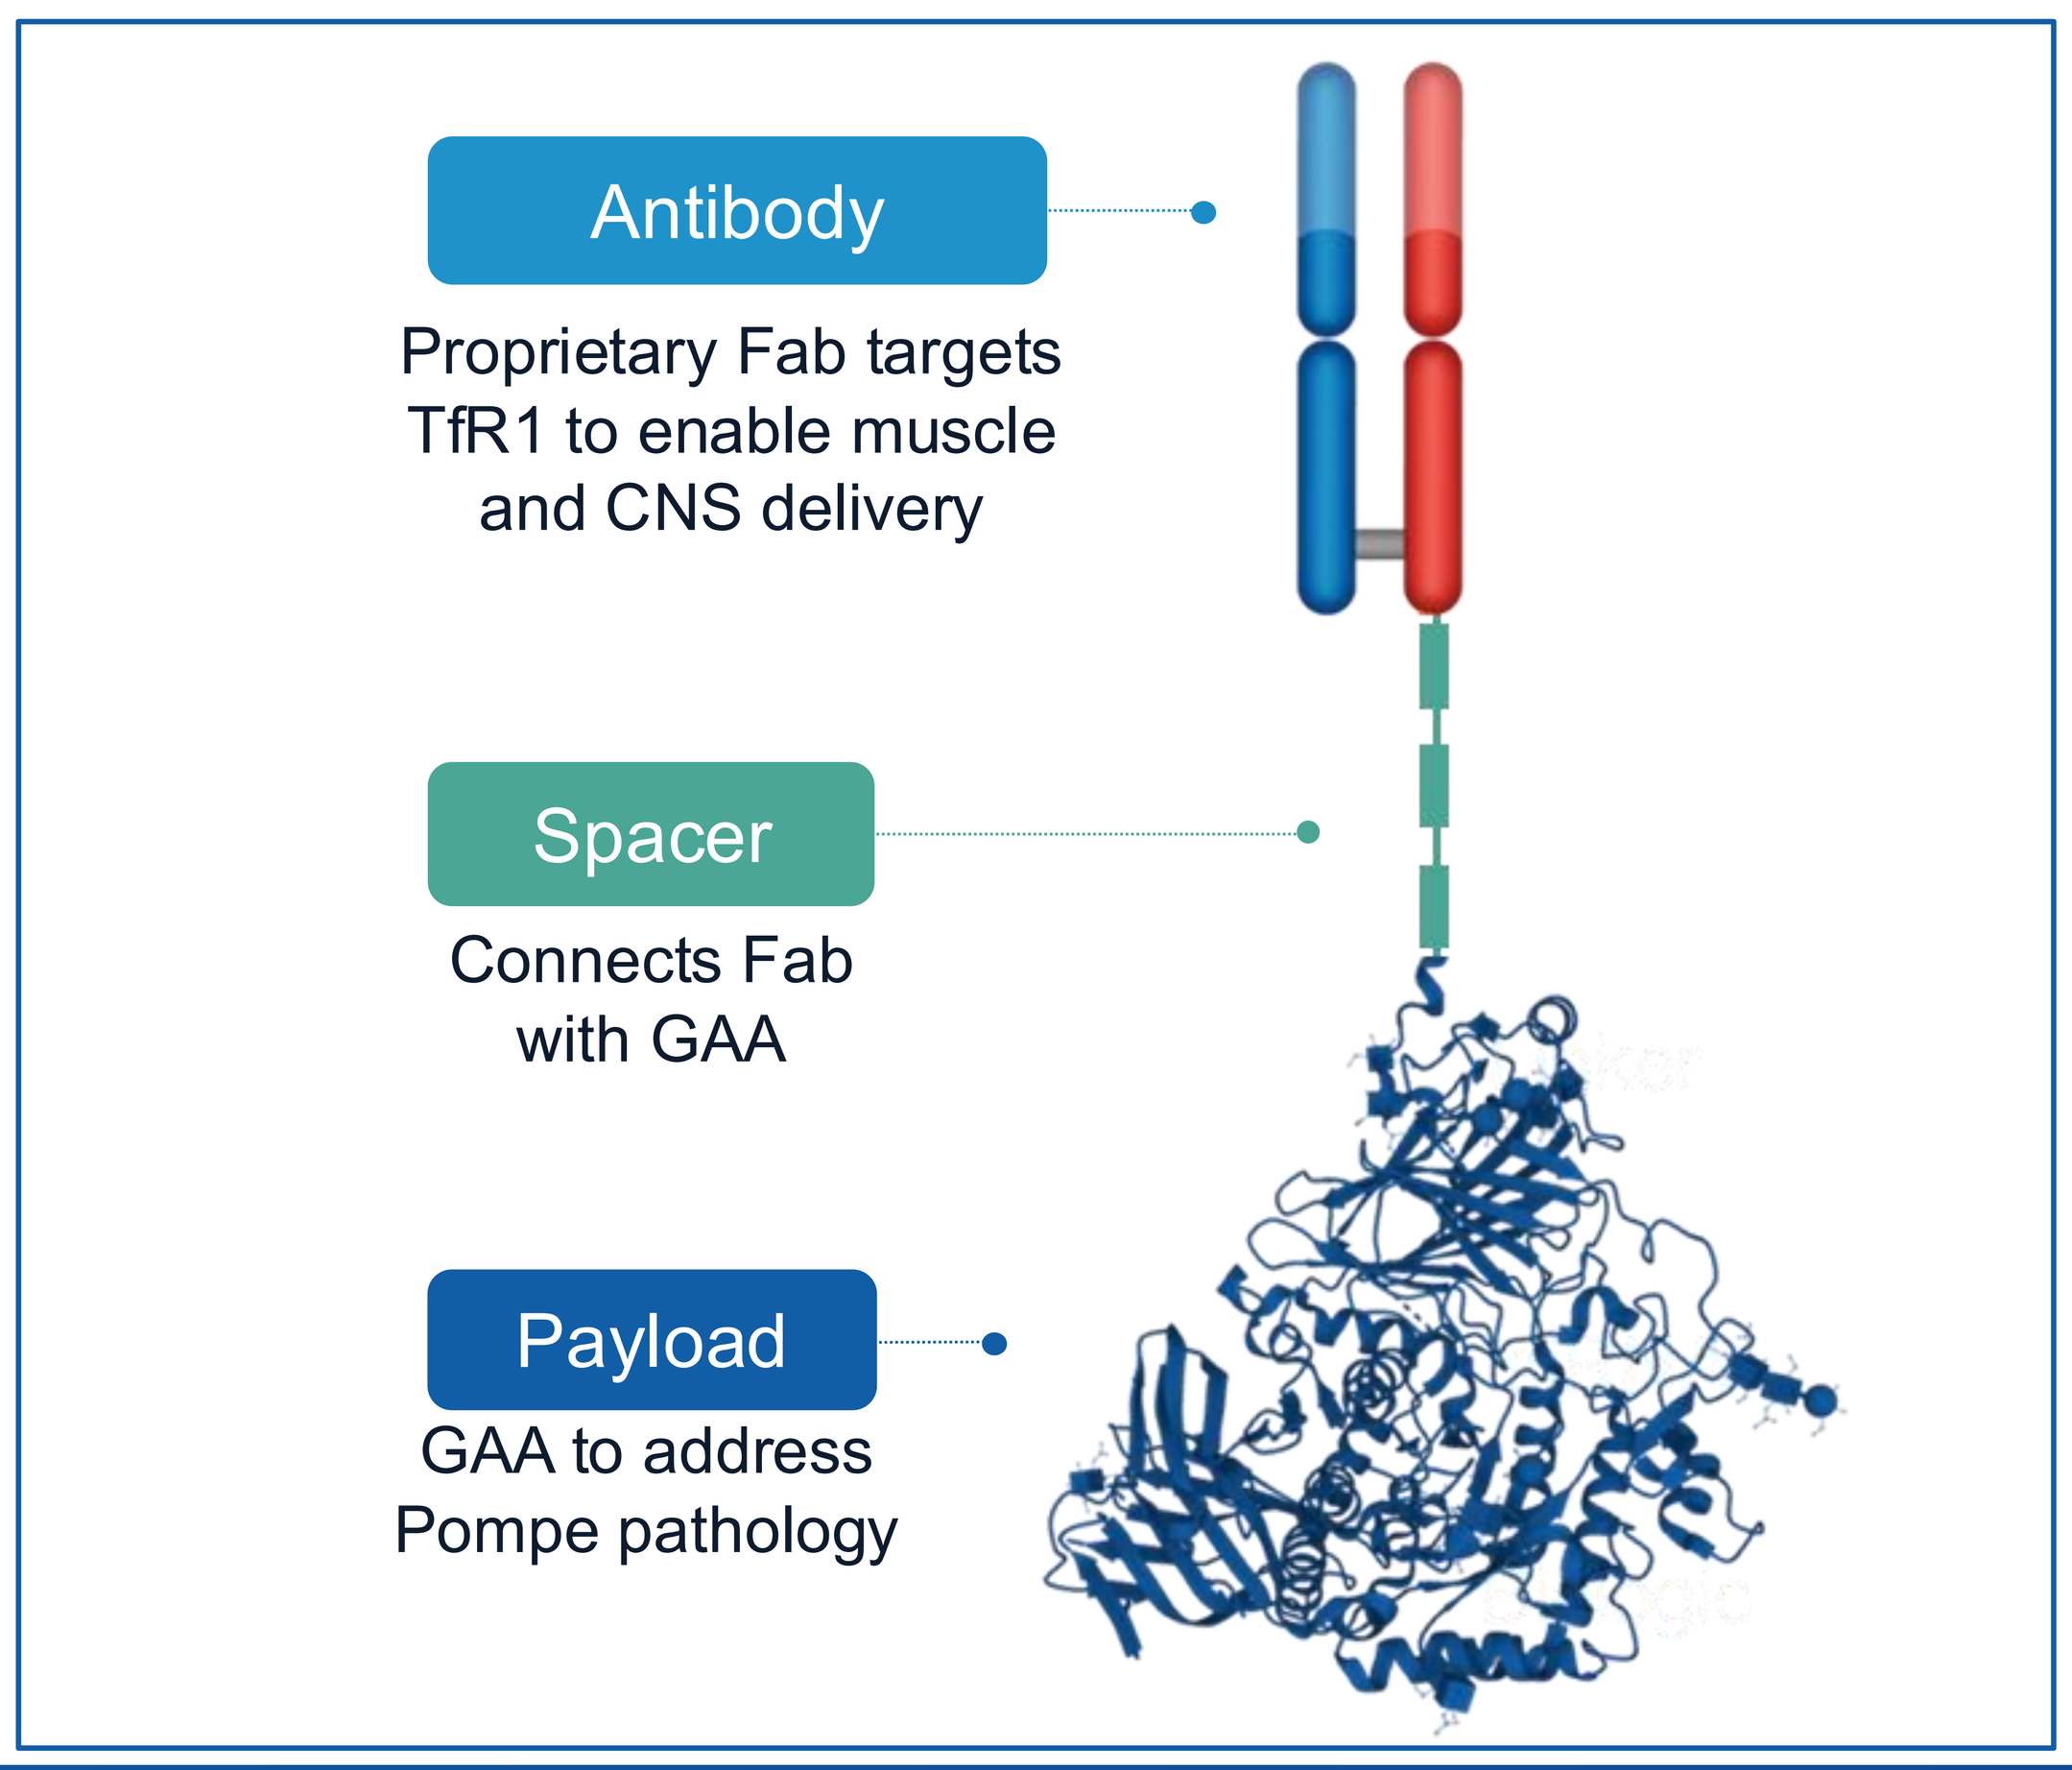

Antibody
Proprietary Fab targets TfR1 to enable muscle and CNS delivery
Spacer
Connects Fab with GAA
Payload
GAA to address Pompe pathology

Figure 2. FORCE-GAA Delivers to the Endo-lysosomal System

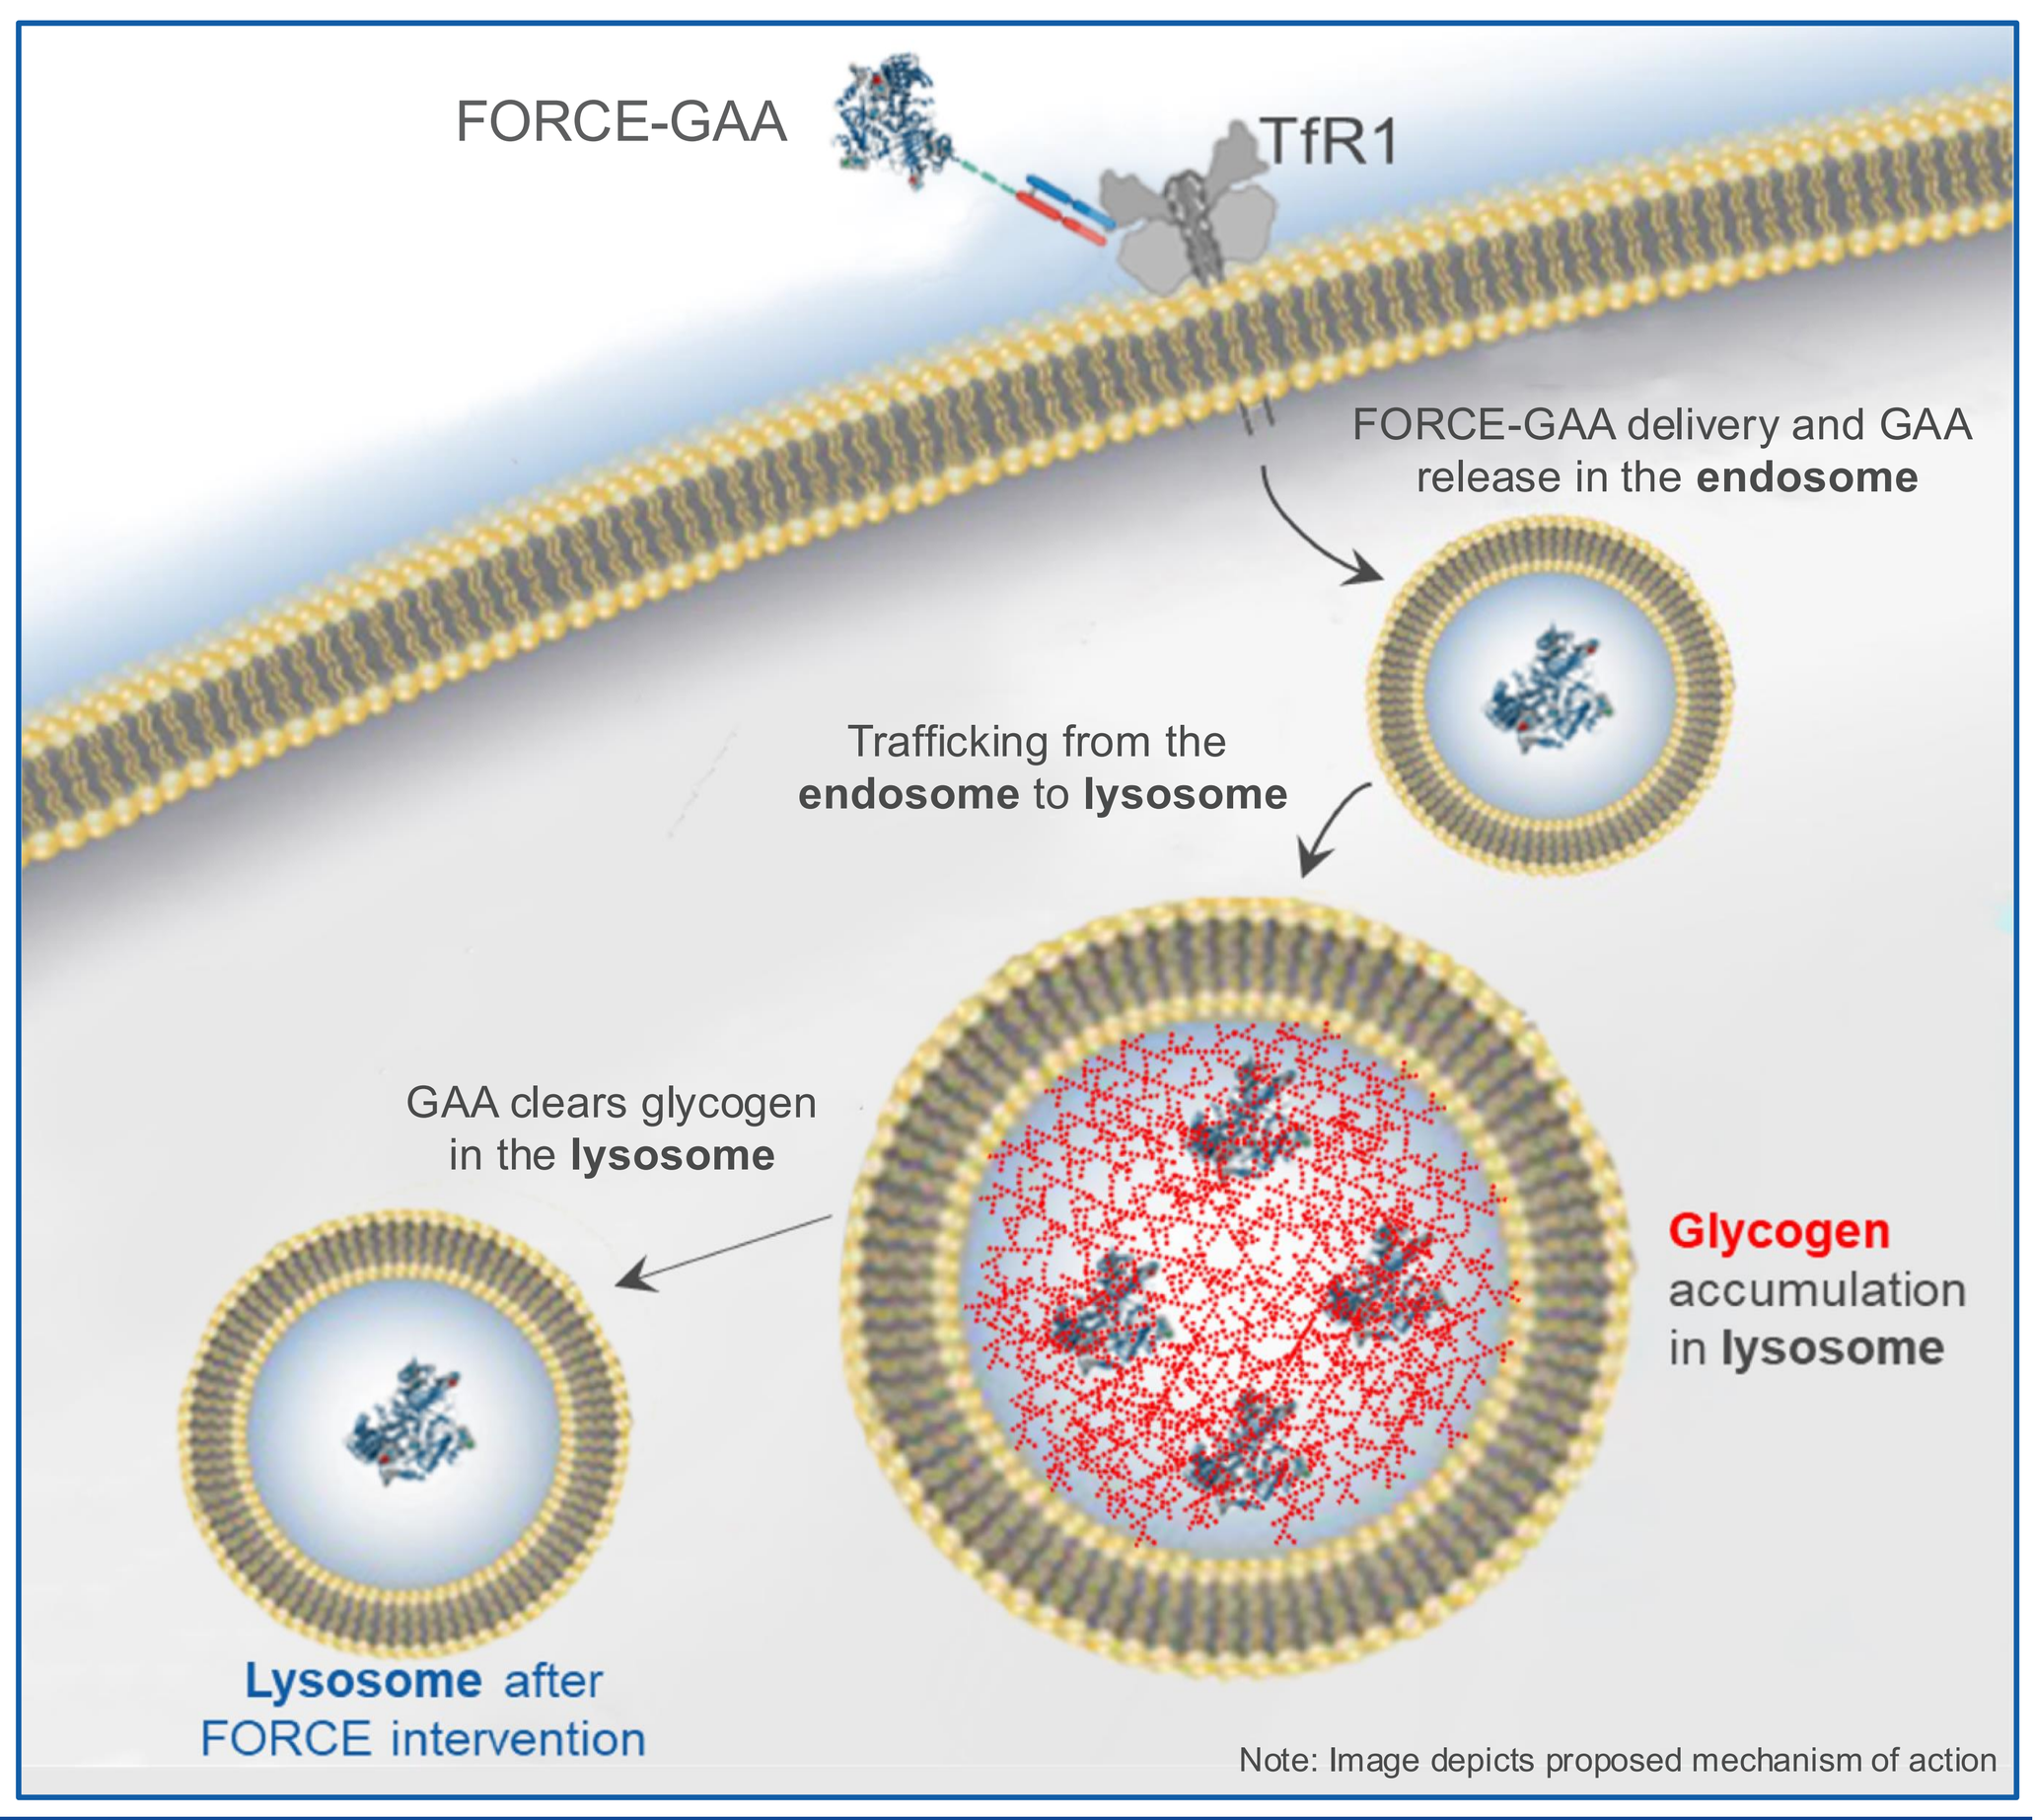

FORCE-GAA
TfR1
FORCE-GAA delivery and GAA release in the endosome
Trafficking from the endosome to lysosome
GAA clears glycogen in the lysosome
Glycogen accumulation in lysosome
Lysosome after FORCE intervention
Note: Image depicts proposed mechanism of action

RESULTS

Figure 3. FORCE-GAA Achieves Superior Glycogen Clearance in Muscle Compared to Naked GAA Using the SOC Dosing Regimen

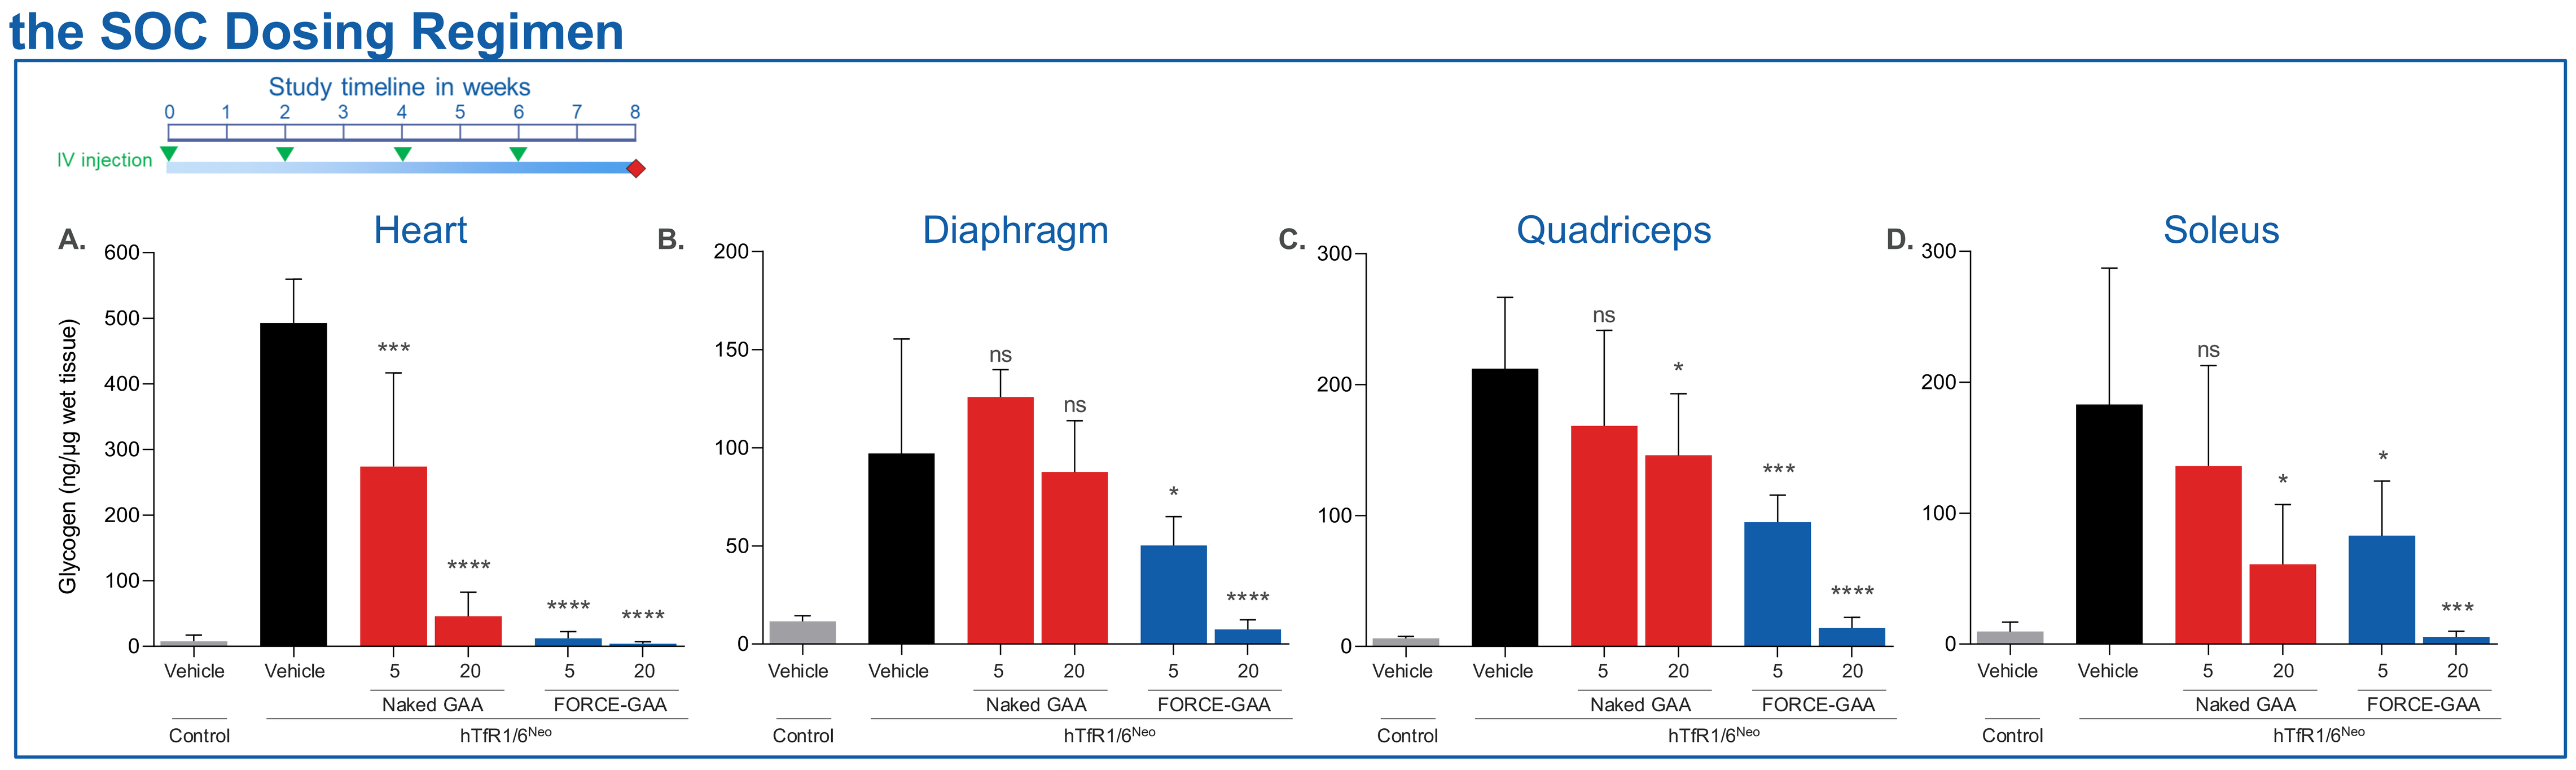

Study timeline in weeks
IV injection
A. Heart
B. Diaphragm
C. Quadriceps
D. Soleus
Glycogen (ng/µg wet tissue)
Vehicle
Naked GAA
FORCE-GAA
Control
hTfR1/6Neo

(A-D) Total glycogen levels in muscle demonstrate superiority of FORCE-GAA over naked GAA in hTfR1/6^{Neo} Pompe disease mice. Doses are mg/kg GAA equivalents. Mice were dosed on day 0 and weeks 2, 4, and 6, analyzed on week 8. Data are means + SD; n=4-7. Control mice are hTfR1(Het)/6^{Neo}(Het); hTfR1/6^{Neo} mice are hTfR1(Het)/6^{Neo}(Hom); Statistical significance compared to vehicle treated hTfR1/6^{Neo} mice by ANOVA $^{*}p<0.05$; $^{***}p<0.001$; $^{****}p<0.0001$. Glycogen levels from tissue homogenates were measured biochemically using a commercially available kit.

Figure 4. FORCE-GAA Demonstrates Superior Reduction of Lysosomal Enlargement Compared to Naked GAA in Muscle Using SOC Dosing

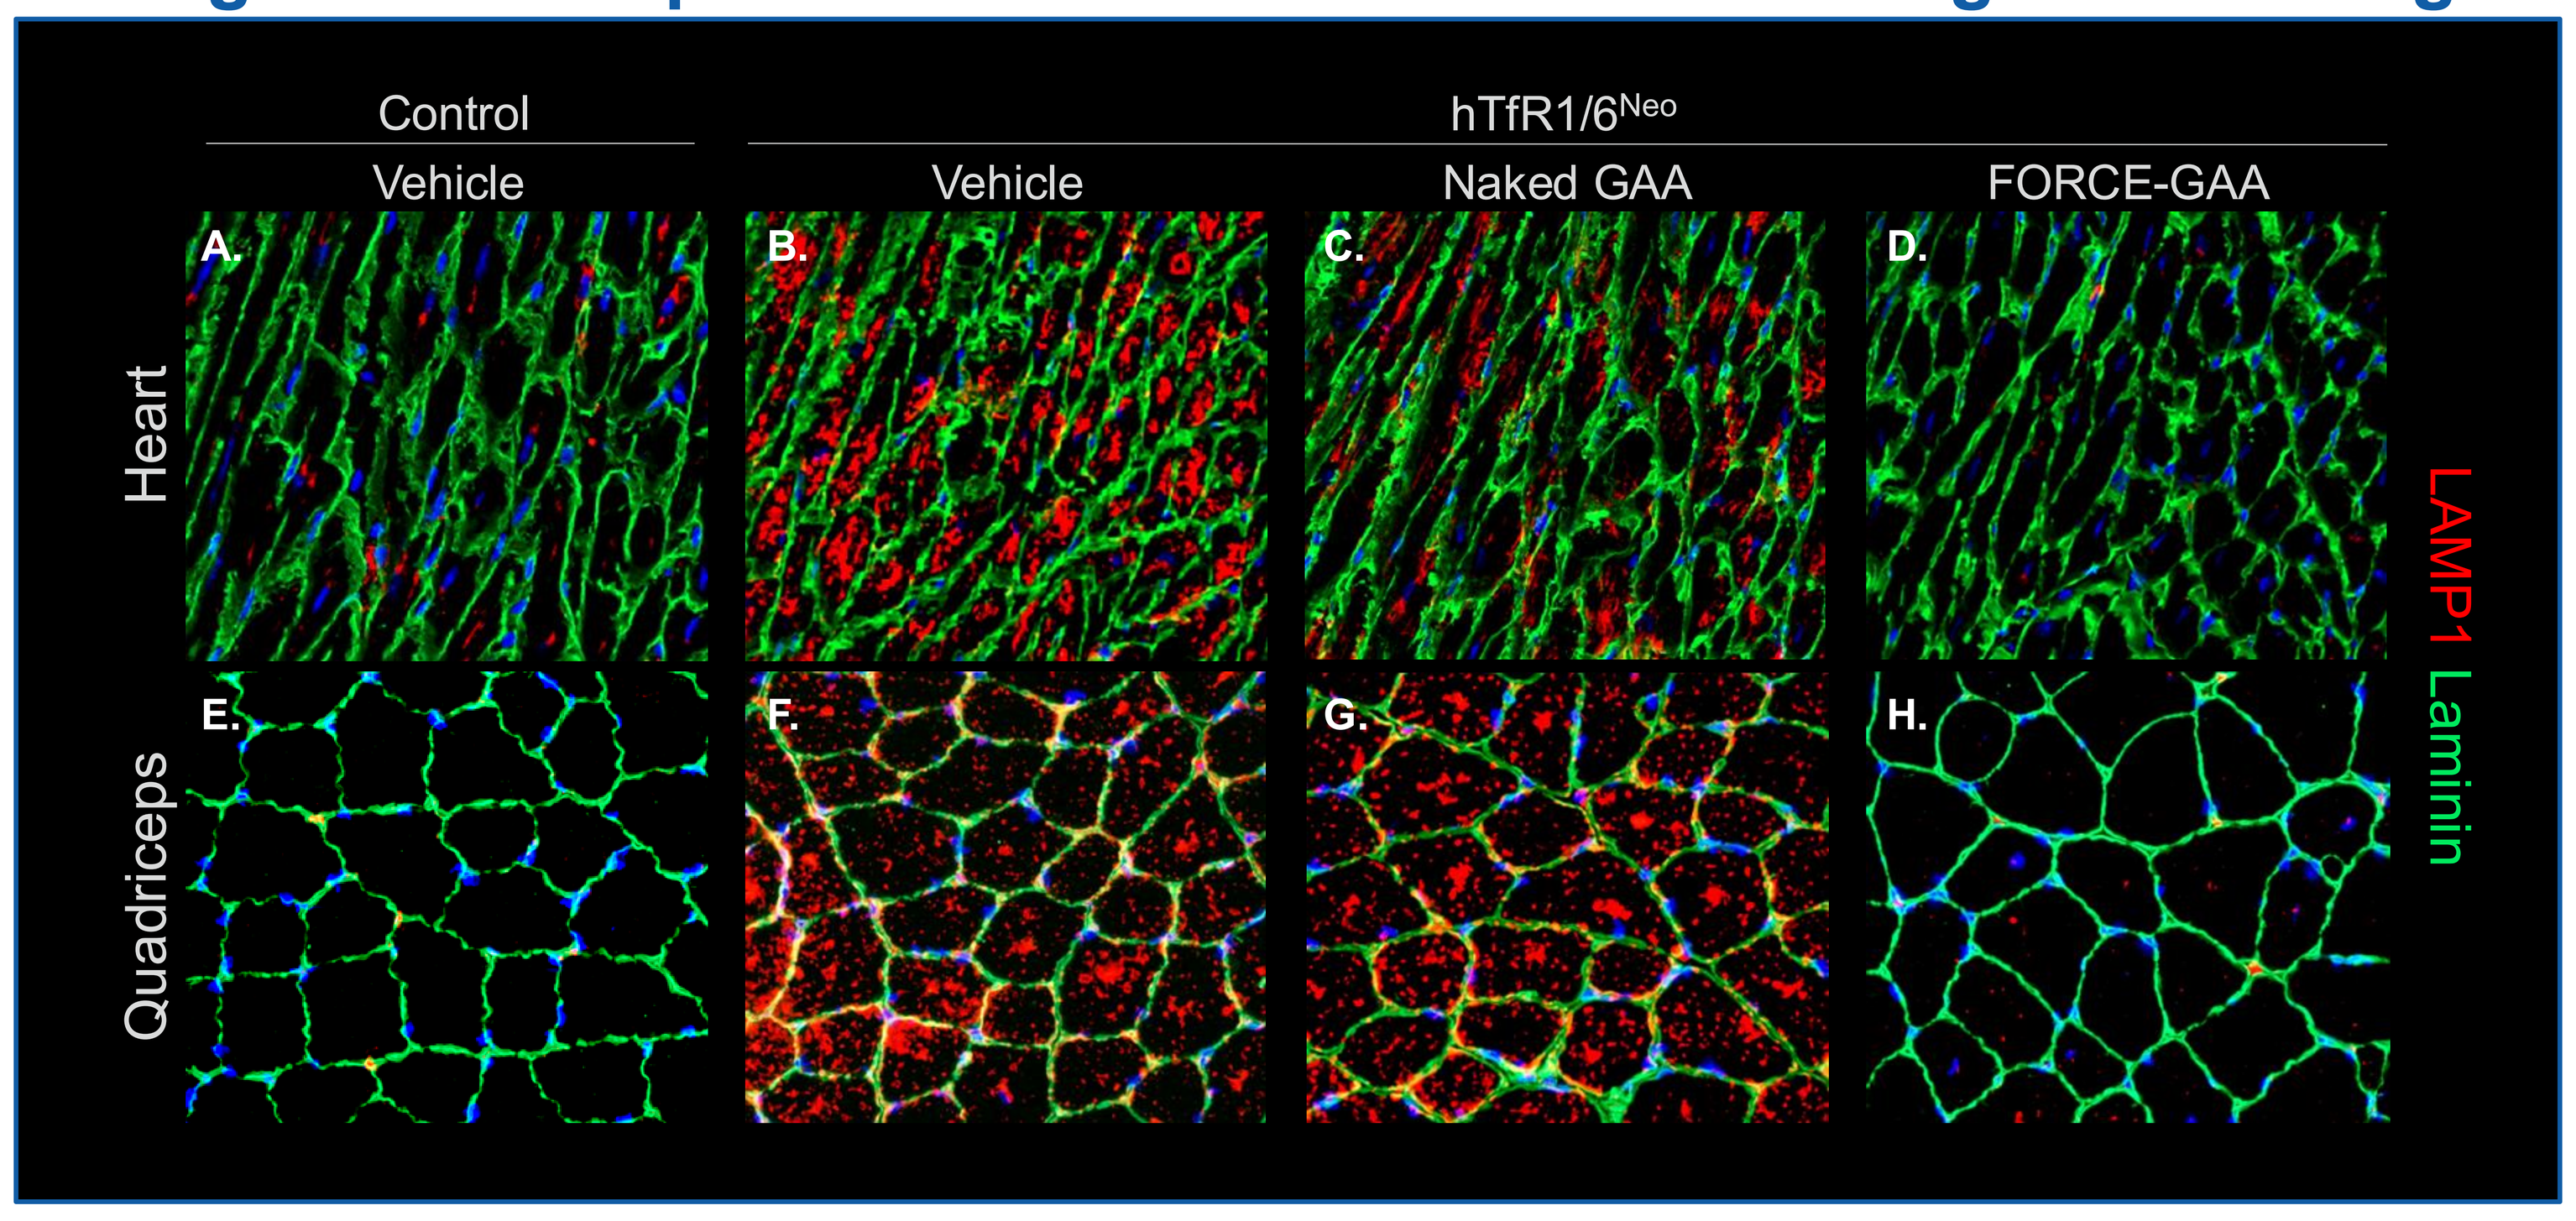

Control
Vehicle
hTfR1/6Neo
Vehicle
Naked GAA
FORCE-GAA
Heart
Quadriceps
LAMP1 Laminin

Representative images of lysosomal enlargement in heart (A-D) and quadriceps (E-H). Superior reduction of lysosomal enlargement in muscle is observed in FORCE-GAA treated hTfR1/6^{Neo} Pompe disease mice. Dose is 20 mg/kg GAA-equivalents. Mice were dosed on day 0 and weeks 2, 4, and 6, analyzed on week 8. Control mice are hTfR1(Het)/6^{Neo}(Het); hTfR1/6^{Neo} mice are hTfR1(Het)/6^{Neo}(Hom). Frozen sections were processed and tissue sections were stained with antibodies against Lysosome associated membrane protein 1 (LAMP1) and Laminin to visualize lysosomes and basement membranes, respectively.

Figure 5. FORCE-GAA Clears Glycogen in the CNS and Normalizes Serum Neurofilament Light Chain (NF-L) Using the SOC Dosing Regimen

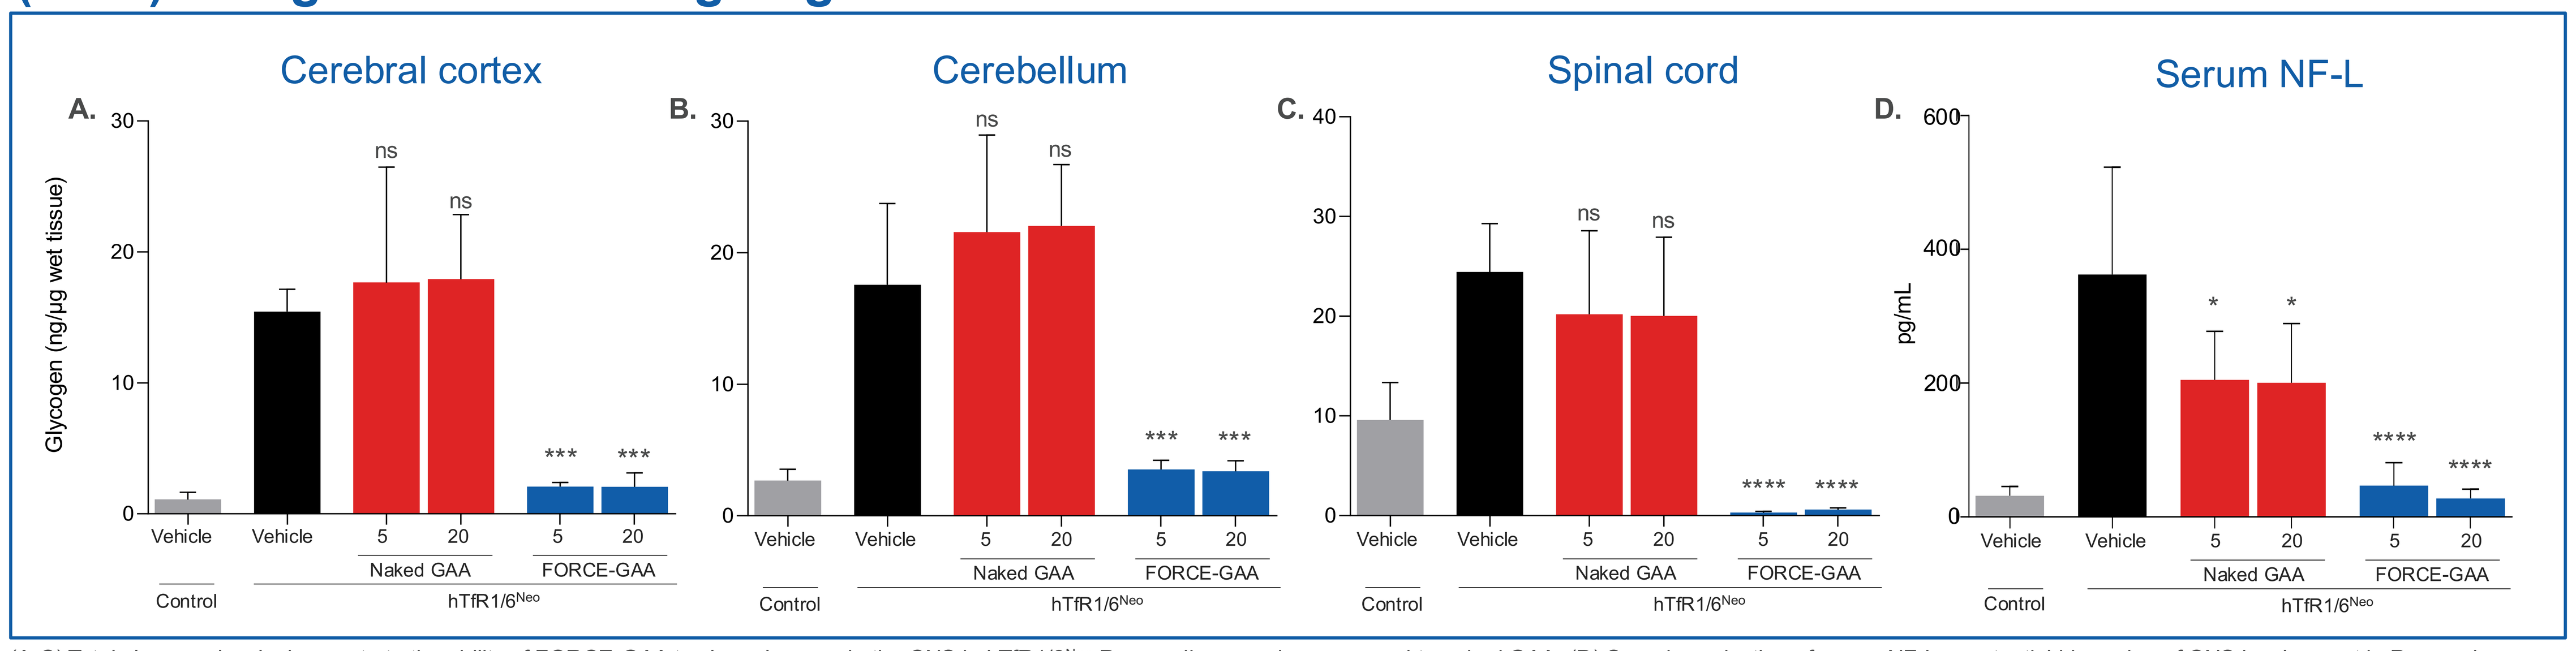

A. Cerebral cortex
B. Cerebellum
C. Spinal cord
D. Serum NF-L
Glycogen (ng/µg wet tissue)
pg/mL
Vehicle
Naked GAA
FORCE-GAA
Control
hTfR1/6Neo

(A-C) Total glycogen levels demonstrate the ability of FORCE-GAA to clear glycogen in the CNS in hTfR1/6^{Neo} Pompe disease mice compared to naked GAA. (D) Superior reduction of serum NF-L, a potential biomarker of CNS involvement in Pompe, is observed with FORCE-GAA compared to naked GAA. Doses are mg/kg GAA-equivalents. Mice were dosed on day 0 and weeks 2, 4, and 6, analyzed on week 8. Data are means + SD; n = 4-7. Control mice are hTfR1(Het)/6^{Neo}(Het); hTfR1/6^{Neo} mice are hTfR1(Het)/6^{Neo}(Hom); Statistical significance compared to vehicle treated hTfR1/6^{Neo} mice by ANOVA $^{*}p<0.1$; $^{***}p<0.001$; $^{****}p<0.0001$. Glycogen levels from tissue homogenates were measured biochemically using a commercially available kit.

Figure 6. FORCE-GAA Achieves Widespread Lysosomal Size Normalization in CNS Using SOC Dosing

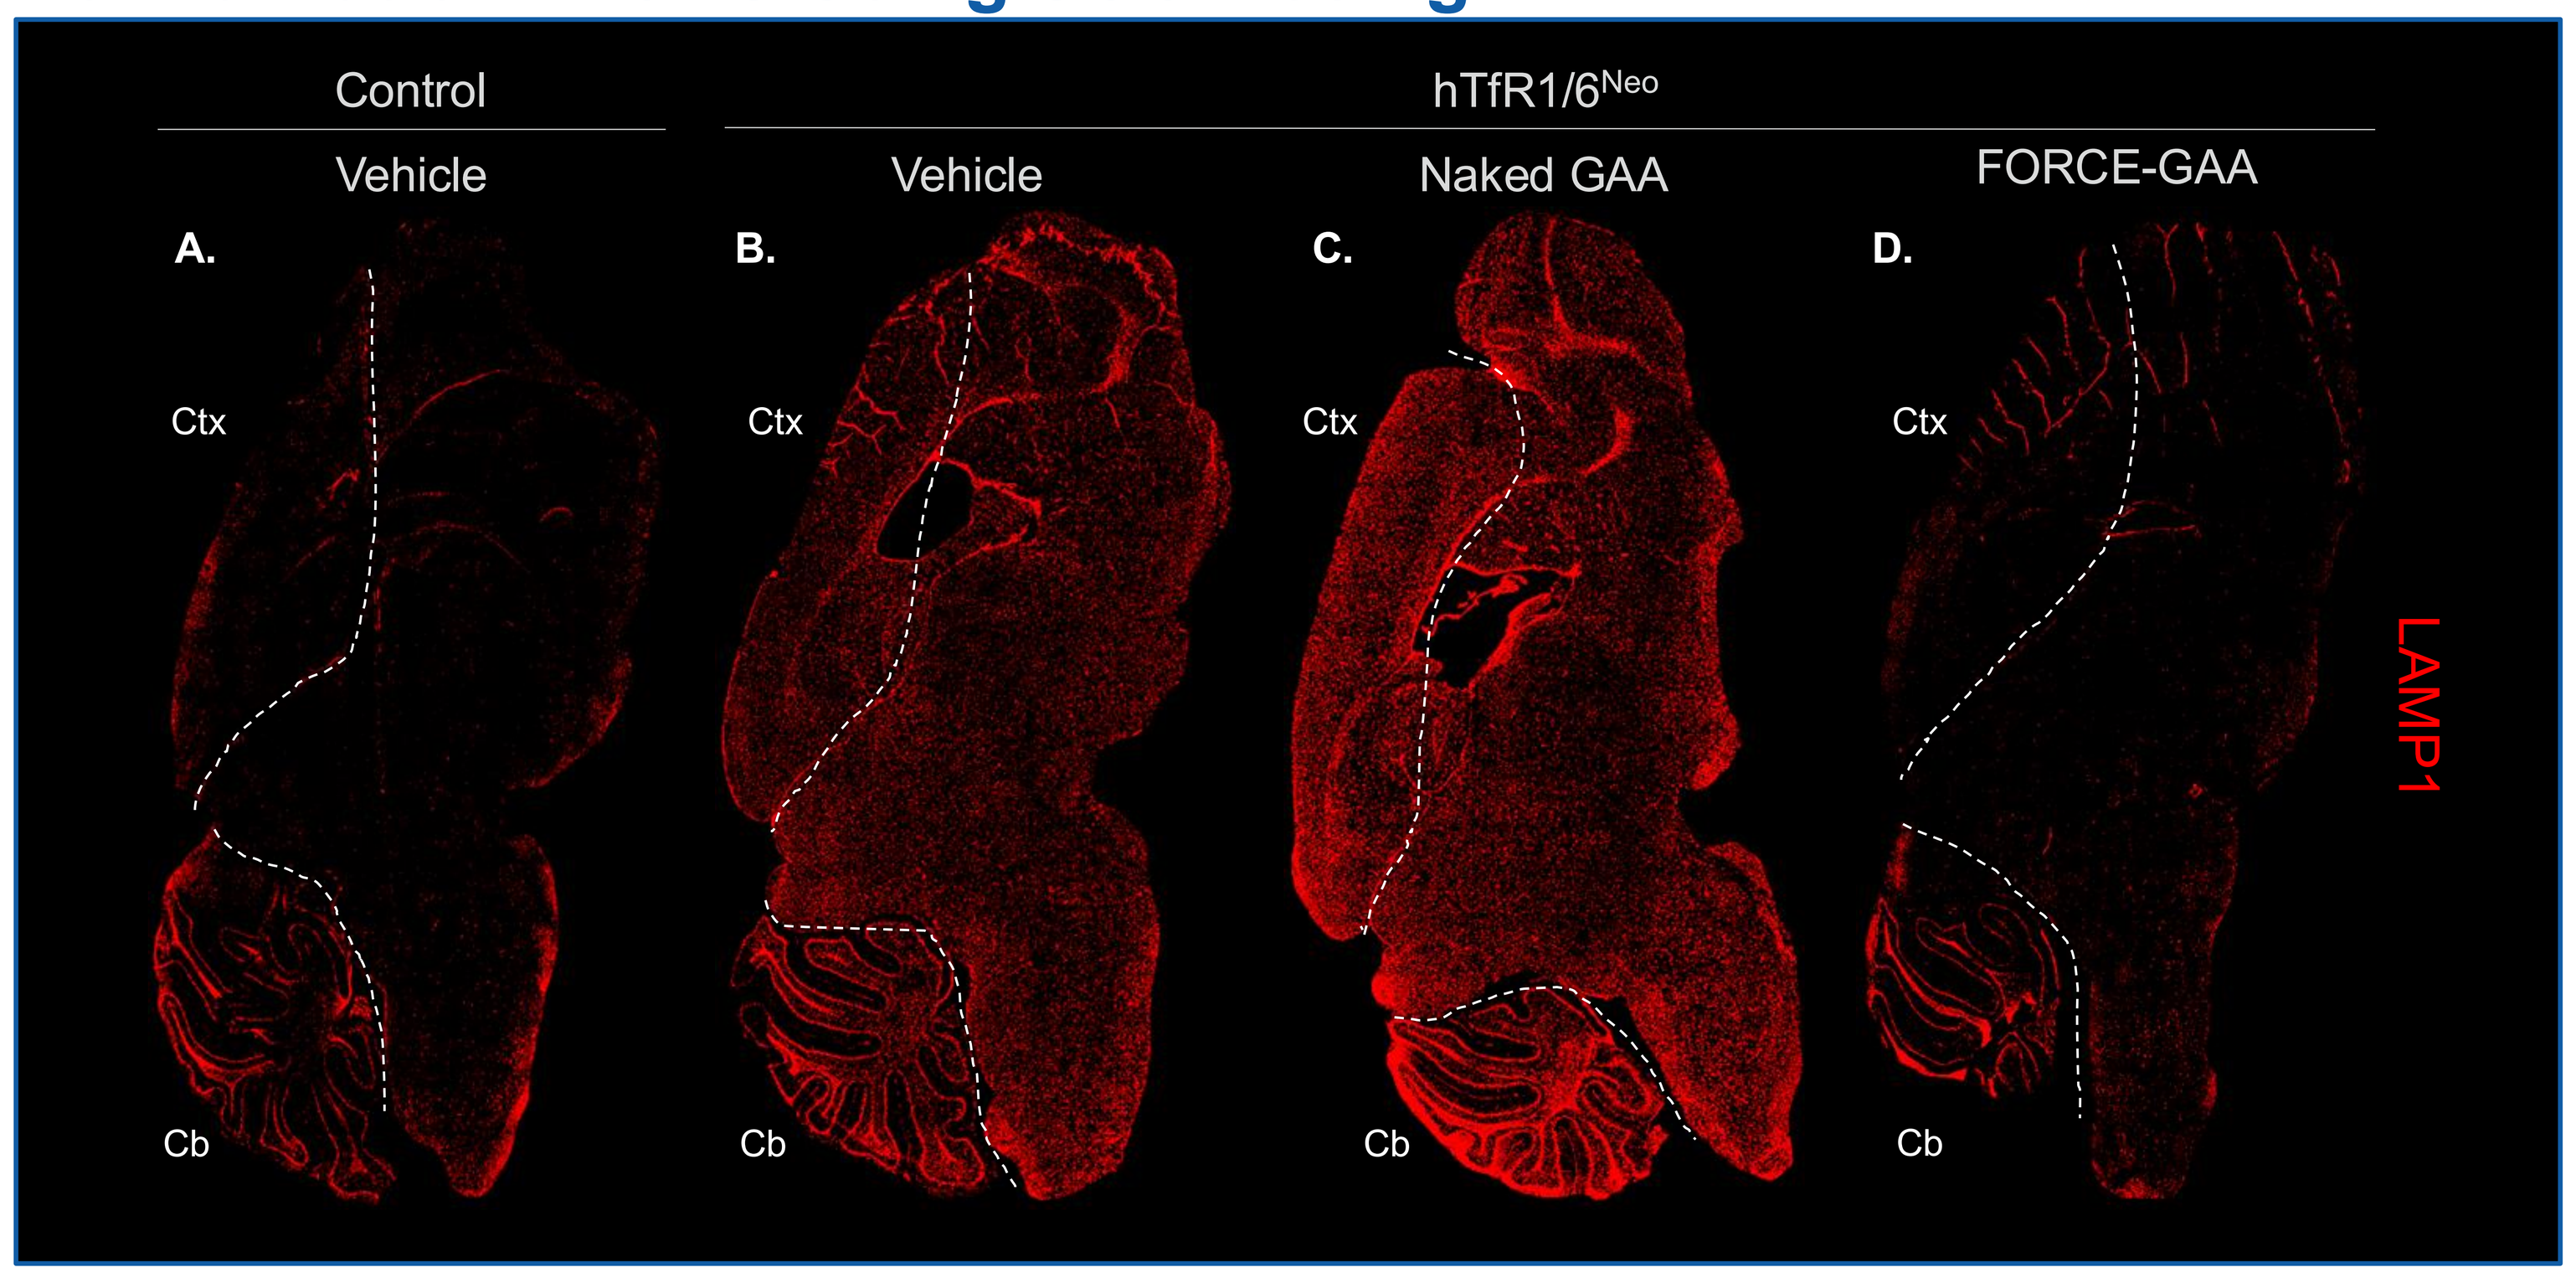

Control
Vehicle
hTfR1/6Neo
Vehicle
Naked GAA
FORCE-GAA
Ctx
Cb
LAMP1

Representative images of lysosomal enlargement in the CNS. Widespread and profound reduction of lysosomal enlargement is observed in FORCE-GAA treated hTfR1/6^{Neo} Pompe disease mice. Dose is 20 mg/kg GAA-equivalents. Mice were dosed on day 0 and weeks 2, 4, and 6, analyzed on week 8. Control mice are hTfR1(Het)/6^{Neo}(Het); hTfR1/6^{Neo} mice are hTfR1(Het)/6^{Neo}(Hom). Formalin-fixed paraffin embedded brains were processed and sagittal sections were stained with an antibody against Lysosome associated membrane protein 1 (LAMP1) to visualize lysosomes throughout the CNS.

Figure 7. FORCE-GAA Monthly Dosing Clears Glycogen in Muscle and CNS and Normalizes Serum NF-L Levels

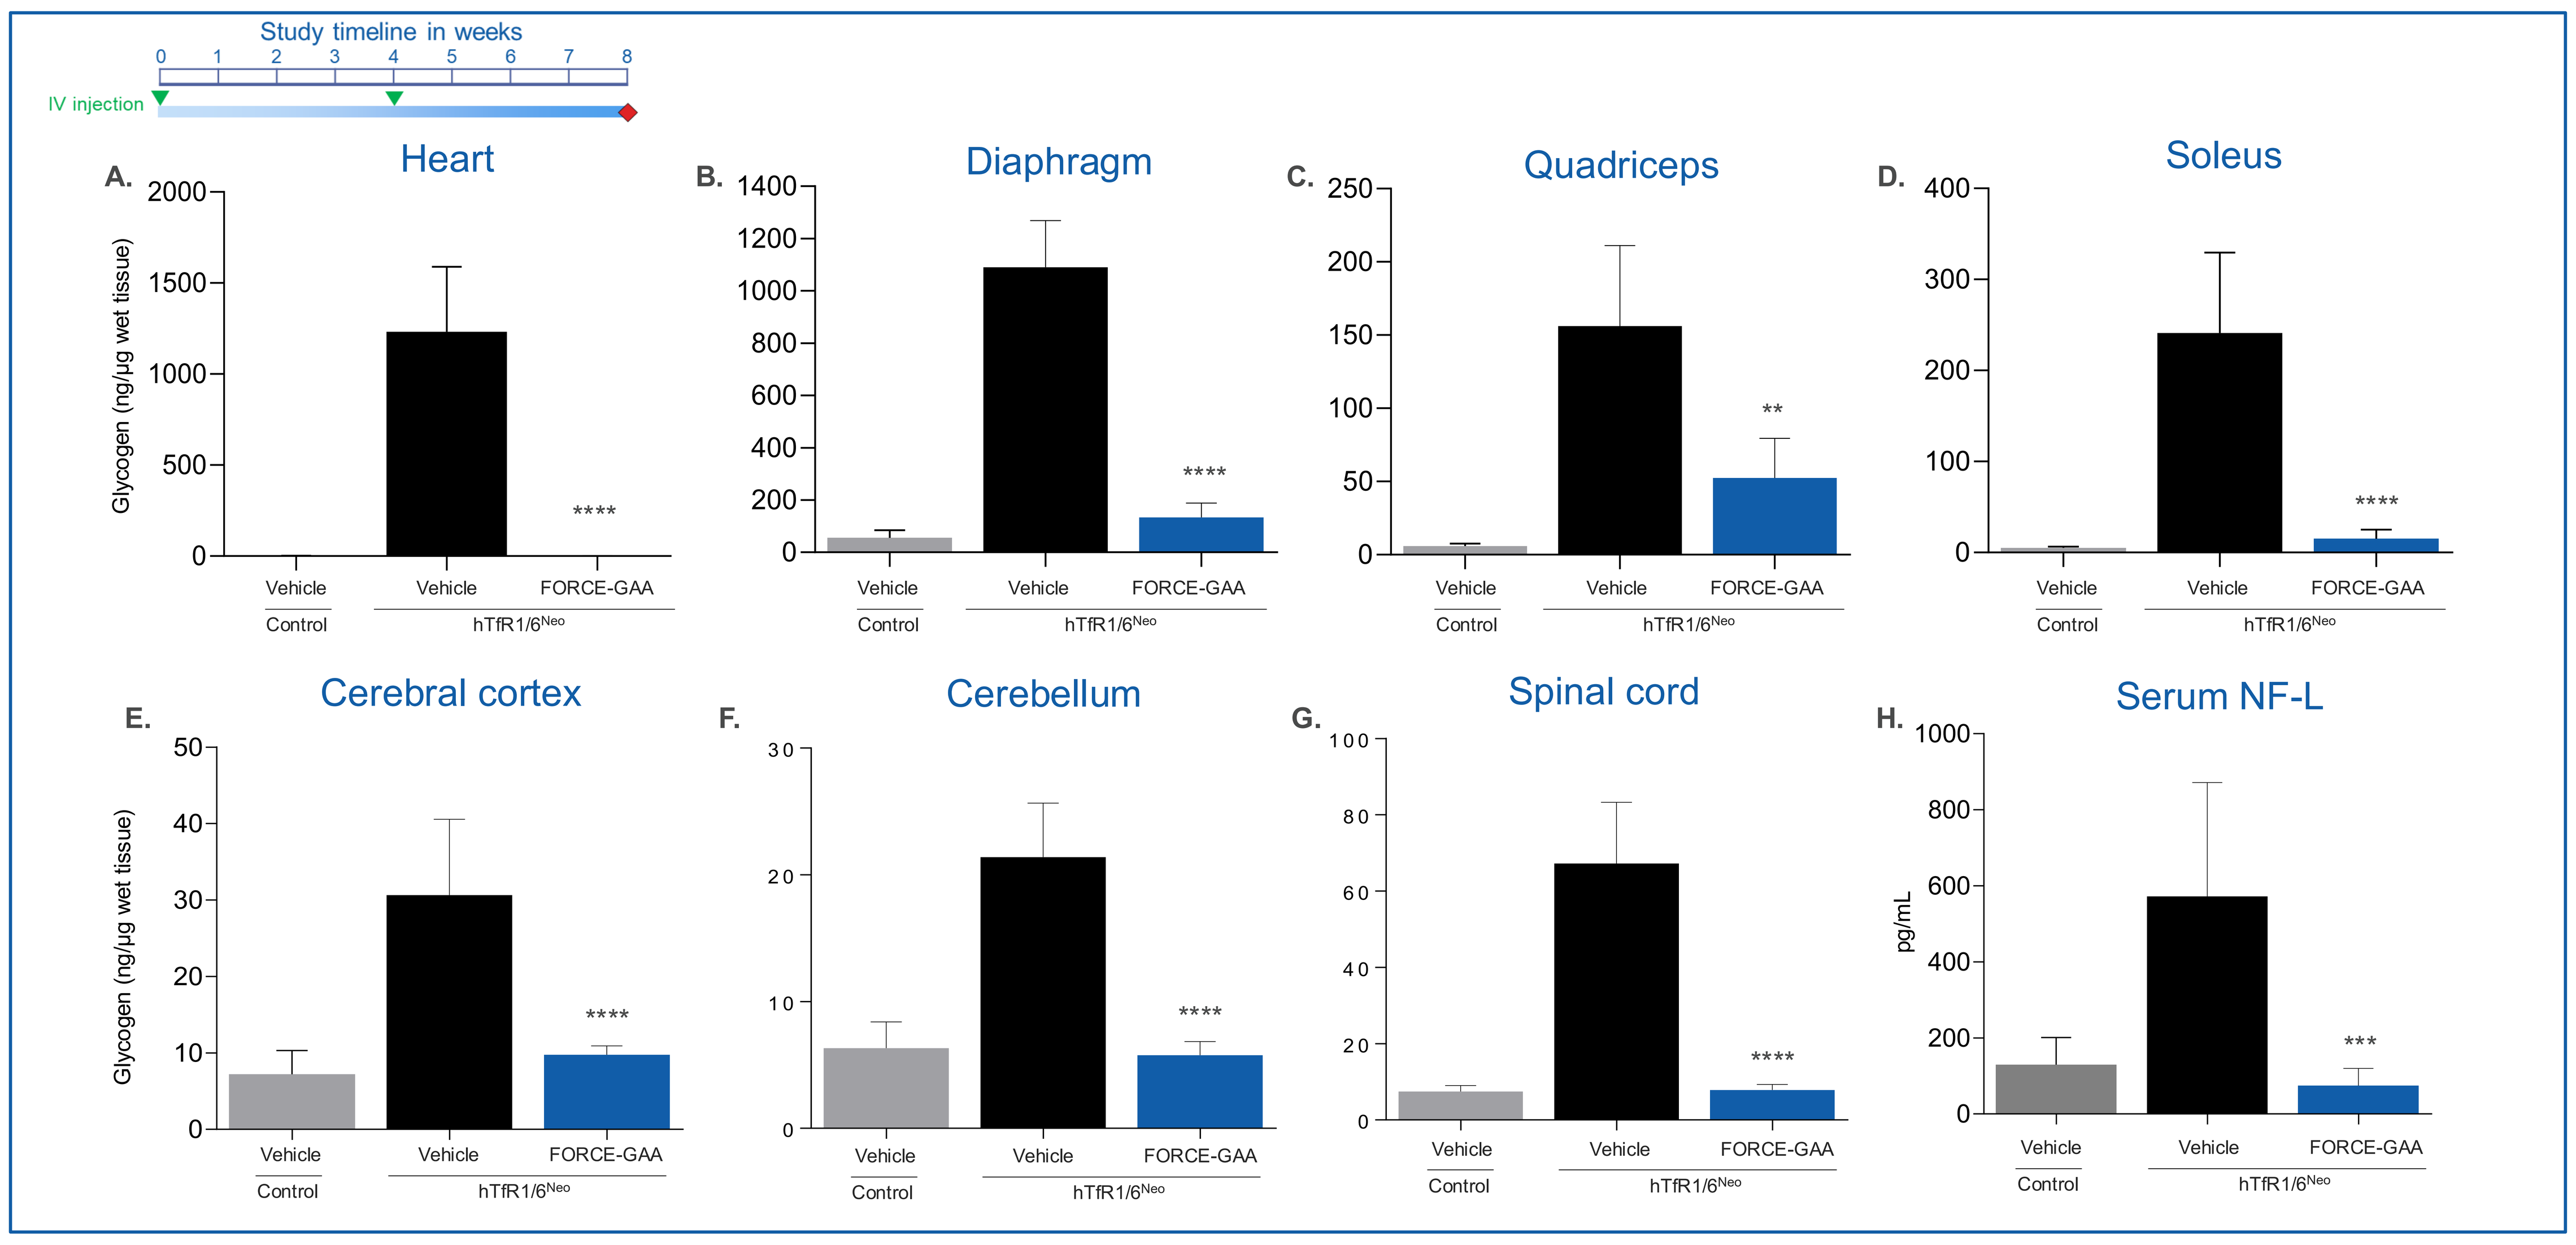

Study timeline in weeks
IV injection
A. Heart
B. Diaphragm
C. Quadriceps
D. Soleus
E. Cerebral cortex
F. Cerebellum
G. Spinal cord
H. Serum NF-L
Glycogen (ng/µg wet tissue)
pg/mL
Vehicle
FORCE-GAA
Control
hTfR1/6Neo

(A-E) Total glycogen levels demonstrate the ability of FORCE-GAA to clear glycogen when dosed Q4W in hTfR1/6^{Neo} Pompe disease. (H) Superior reduction of serum NF-L is observed with monthly dosing of FORCE-GAA. Dose is 20 mg/kg GAA-equivalents. Mice were dosed on day 0 and week 4; analyzed on week 8. Data are means + SD; n = 4-7. Control mice are hTfR1(Het)/6^{Neo}(Het); hTfR1/6^{Neo} mice are hTfR1(Het)/6^{Neo}(Hom); Statistical significance compared to vehicle treated hTfR1/6^{Neo} mice by ANOVA $^{**}p<0.01$; $^{***}p<0.001$; $^{****}p<0.0001$. Glycogen levels from tissue homogenates were measured biochemically using a commercially available kit.

CONCLUSIONS

- FORCE-GAA displayed superior efficacy in cardiac and skeletal muscle compared to naked GAA in a well-established mouse model of Pompe disease
- FORCE enables effective ERT delivery throughout the CNS that translates into normalization of serum NF-L levels in a mouse model of Pompe disease
- Durability of pharmacodynamics in muscle and CNS indicates potential for monthly or less frequent dosing
- Modularity of FORCE as a delivery platform for muscle and CNS is demonstrated with a biologic payload

Data support the applicability of the FORCE platform for the treatment of Pompe

REFERENCES

1. Lim, J-A. *et al., Front Aging Neurosci.*, 2014.
2. Van der Ploeg, A.T. and Reuser, A.J., *Lancet*, 2008
3. Aung-Htut, M.T., *et al., Sci Rep.* 2020
4. Fusco, A.F., *et al., Int. J. Mol. Sci.*, 2020
5. van der Beek, N.AME., *et al., Orphanet J. Rare Dis.*, 2012
6. Al Jasmi, F. *et al., BMC Neurol.*, 2015
7. Baik, A.D., *et al., Molecular Therapy*, , 2021
8. Lim, J-A., *et al., Mol. Ther. Methods Clin. Dev.*, 2019
9. Rabin, N., *et al., Mol. Genet. Metab.*, 2003

DISCLOSURE INFORMATION

All authors are employees or advisors of Dyne Therapeutics Inc. and may hold Dyne stock and/or stock options.

World Muscle Society 8-12 October 2024 Prague, Czechia